NOVADISCOVERY
THE EFFECT MODEL COMPANY

ENYO PHARMA

An *in silico* HBV model predicts viral response to the oral non-steroidal carboxylic acid FXR agonist EYP001a

Claudio Monteiro Sousa (1), Evgueni Jacob (1) , Bastien Martin (1) , Samuel Laheux (1), Diane Sampson (2), Elise Roy (2), Jacky Vonderscher (2), Pietro Scalfaro (2), Patrice André (3), Jean-Pierre Boissel (1)

(1) Novadiscovery, Lyon, France, (2) ENYO pharma, Lyon, France, (3) CIRI - INSERM U1111 – CNRS UMR5308 - Université Lyon 1- ENS de Lyon

BACKGROUND

In line with the guidance of the FDA to expand the use of *in silico* trial simulations to support drug development [1], a mathematical model of hepatitis B virus (HBV) infection was built to simulate the effect of EYP001a treatment on HBV replication.

EYP001a is a synthetic non-bile acid Farnesoid X receptor (FXR) agonist currently under clinical development for chronic HBV infection and NASH. FXR regulates bile acid metabolism and is a target for liver disease therapies. We aimed at exploring the EYP001a effect on hepatocyte viruses and viral markers production.

METHODS

The mechanistic model was based on curated knowledge extracted from white and grey scientific literature via the community-driven knowledge management platform (https://githealth.io).

The complete model (142 ODEs, 359 parameters) integrated bile acids physiology, cholesterol metabolism, HBV replication and compound mode of action (the latter from EYP001a non-clinical data, Fig. 1).

The computational model was written and implemented through Novadiscovery's proprietary simulation framework and its various tools (SimWork). The SimWork virtual population and exploration tools were used to calibrate the model: 1,000 virtual patients were randomly generated from ranges of descriptor (representing the n model parameters) values and were selected on the basis of a score translating physiological and biological constraints that the model should comply with; this results in a n-dimension space domain where the parameter values meet the constraints. Qualitative model validation was done through comparison of simulation outputs to known biological systems/disease dynamics.

An independent team, blinded to available clinical EYP001a data, simulated the effect of single and multiple oral doses of EYP001a in healthy or chronically infected HBV virtual subjects. EYP001a effects on FXR response markers 7α-Hydroxy-4-cholesten-3-one (C4) and Fibroblast growth factor 19 (FGF19) were explored. Model performance was tuned with data observed in healthy subjects and simulated results will be quantitatively validated with data from both in vitro HepaRG experiments and *in vivo* HBV infected subjects. The effect on HBV replication of several combinations of EYP001a dosing regimens was explored. Additionally, different associated daily dietary intakes of cholesterol schemes were tested. HBV DNA output curve was generated for 1,000 virtual HBV subjects treated for 100 days with EYP001a.

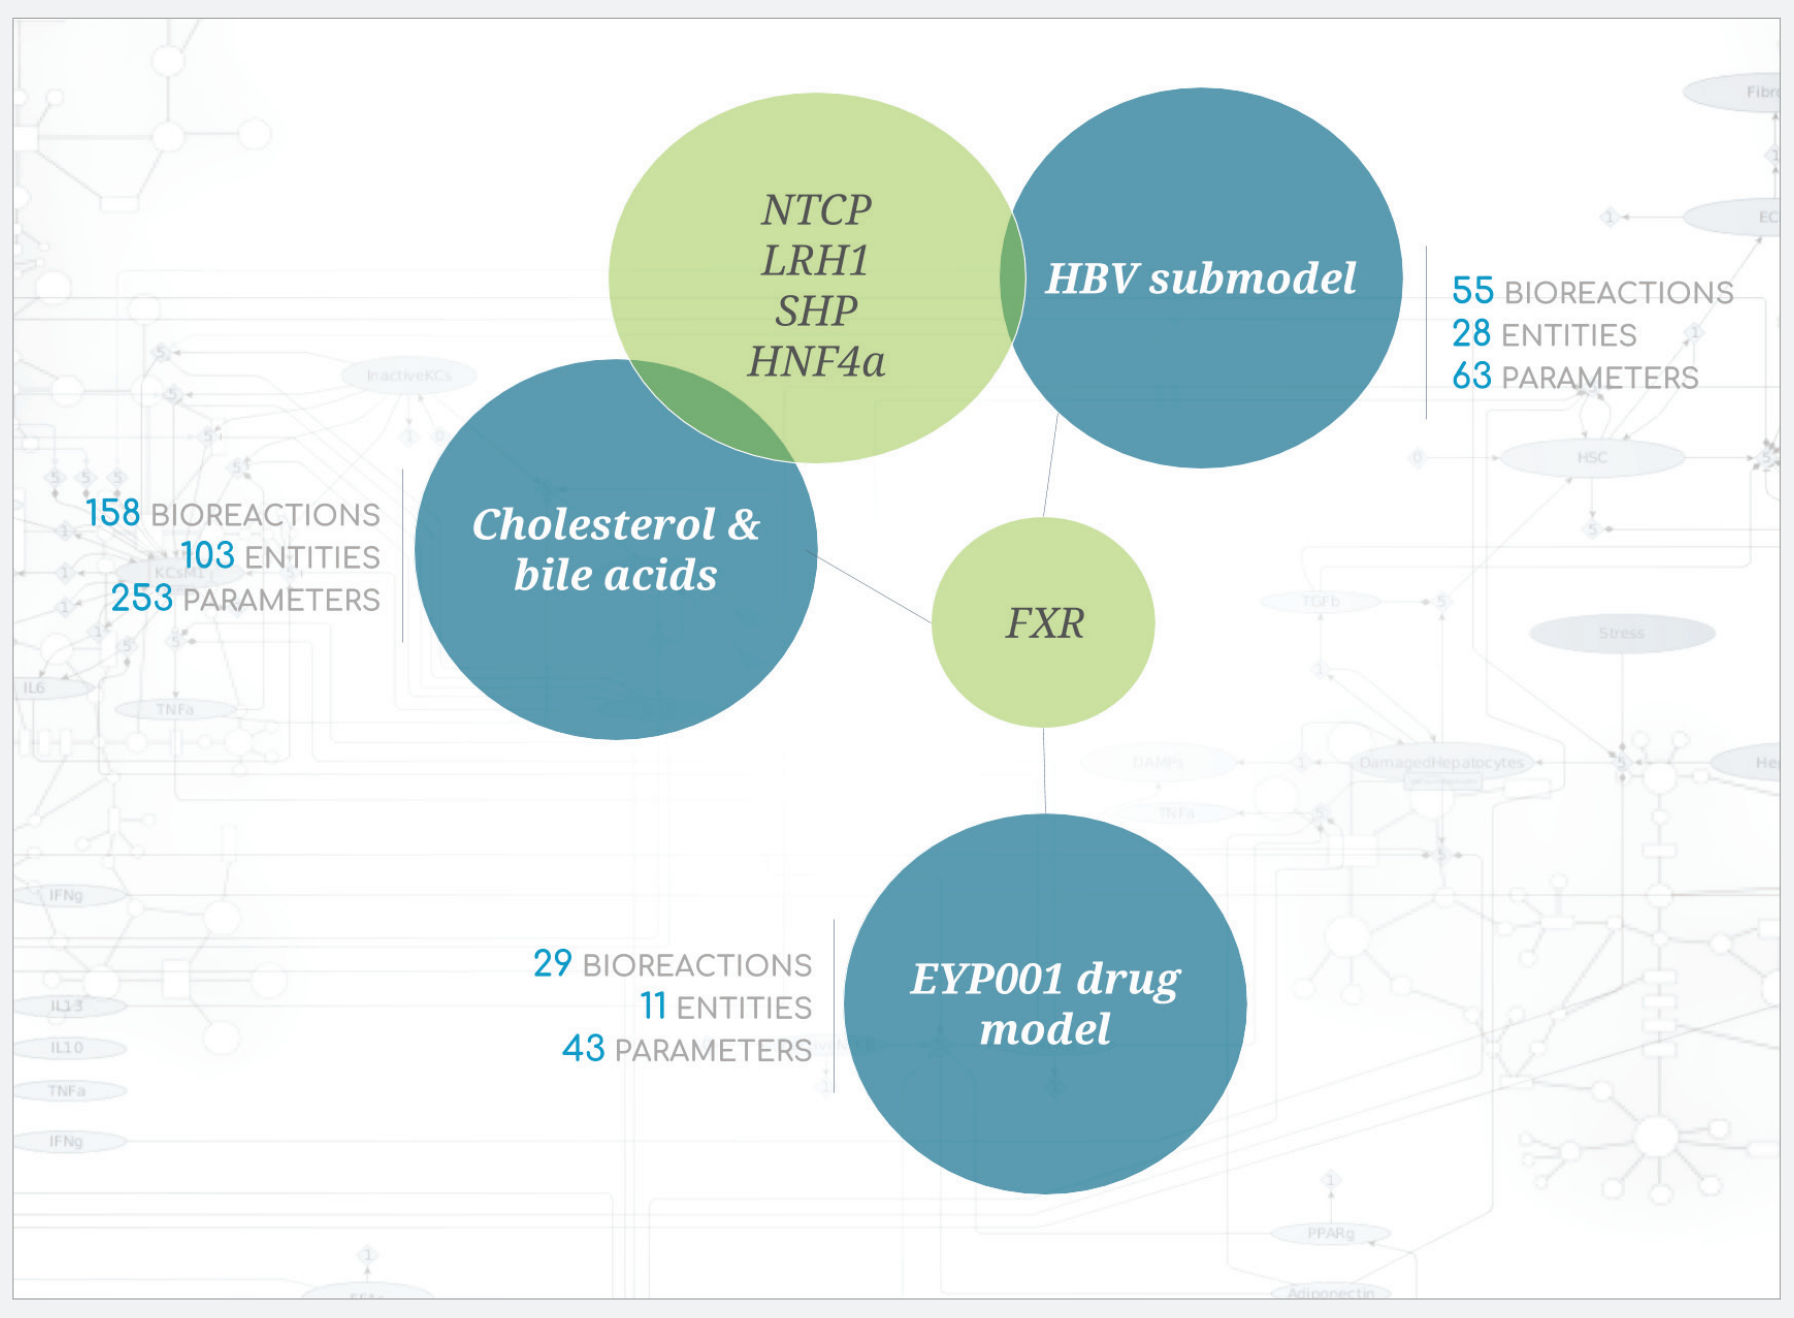
NTCP
LRH1
SHP
HNF4a
HBV submodel
55 BIOREACTIONS
28 ENTITIES
63 PARAMETERS
158 BIOREACTIONS
103 ENTITIES
253 PARAMETERS
Cholesterol & bile acids
FXR
29 BIOREACTIONS
11 ENTITIES
43 PARAMETERS
EYP001 drug model

Figure 1: Computational model structure. Are presented in green entities used as connectors for the different submodels (in blue).

RESULTS

The model successfully reproduced EYP001a plasma concentration-time profiles (Cmax, Tmax and AUC) at the different tested doses (Fig. 2).

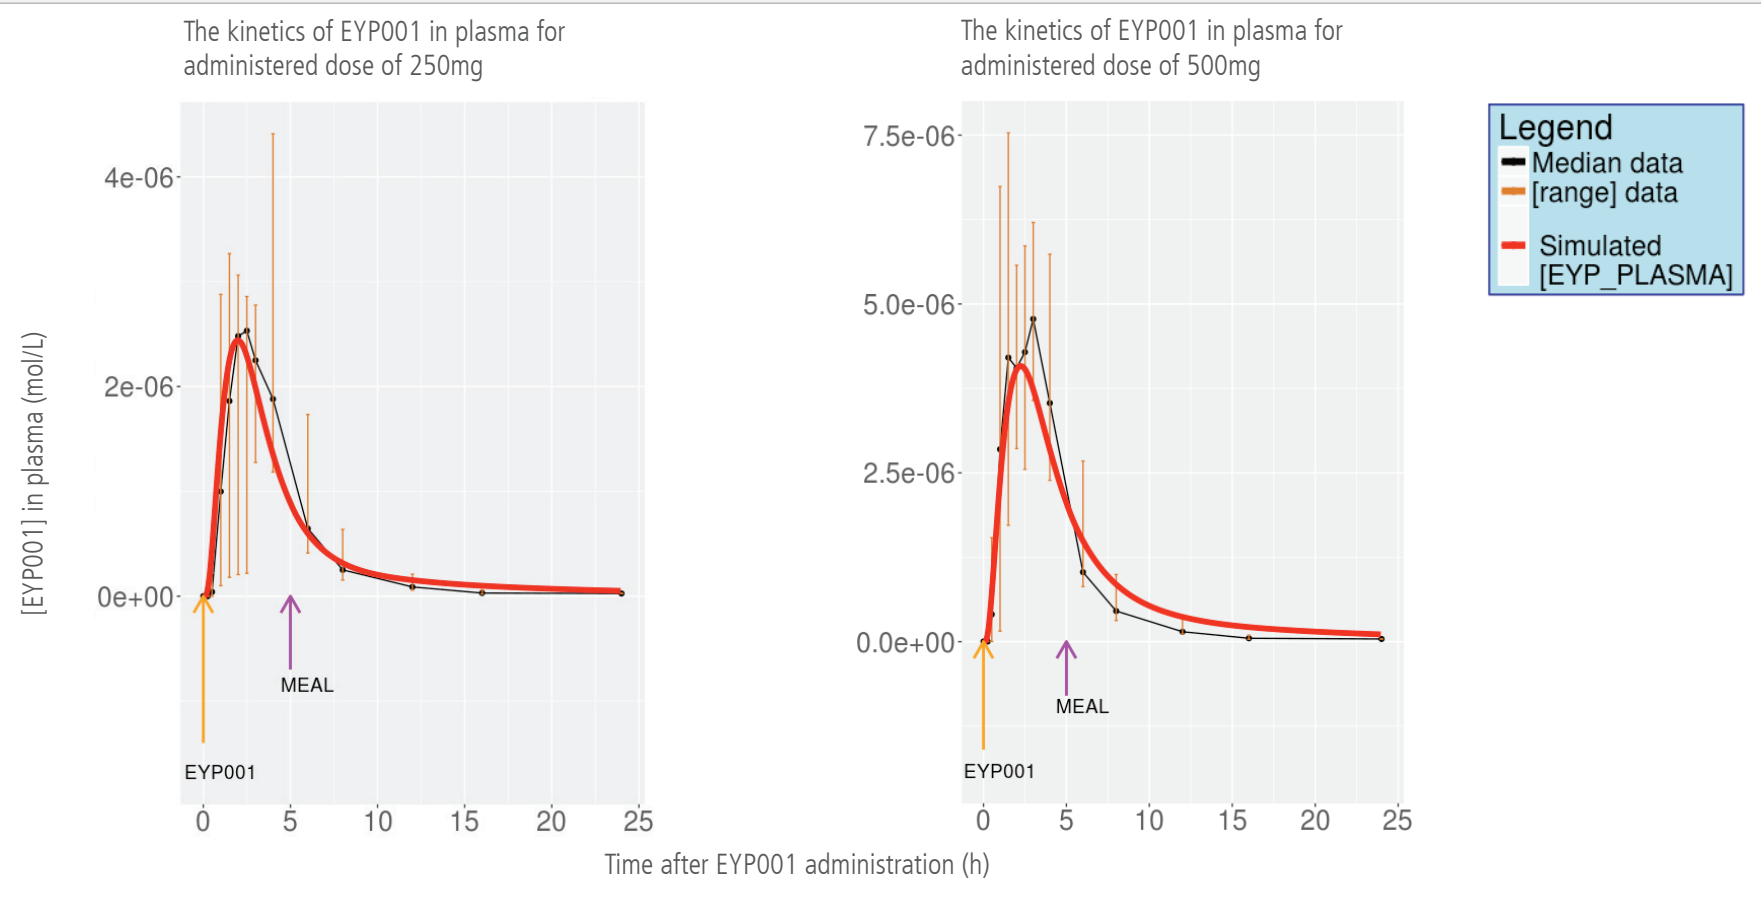
The kinetics of EYP001 in plasma for administered dose of 250mg
The kinetics of EYP001 in plasma for administered dose of 500mg
[EYP001] in plasma (mol/L)
Time after EYP001 administration (h)
EYP001
MEAL
Legend
Median data
[range] data
Simulated [EYP_PLASMA]

Figure 2: kinetics of EYP001a in plasma for the administered doses of 250mg (on the left) and 500mg (on the right). 30mg, 60mg, 120mg and 800mg doses were also successfully reproduced (not presented). The black dots and the brown bars represent respectively the median and the minimum-maximum range of experimental data. Red solid lines correspond to the EYP001a simulated concentrations.

The model reproduced accurately the dynamics of C4 and FGF19 and their changes after single and multiple EYP001a administrations (Fig. 3a and Fig. 3b).

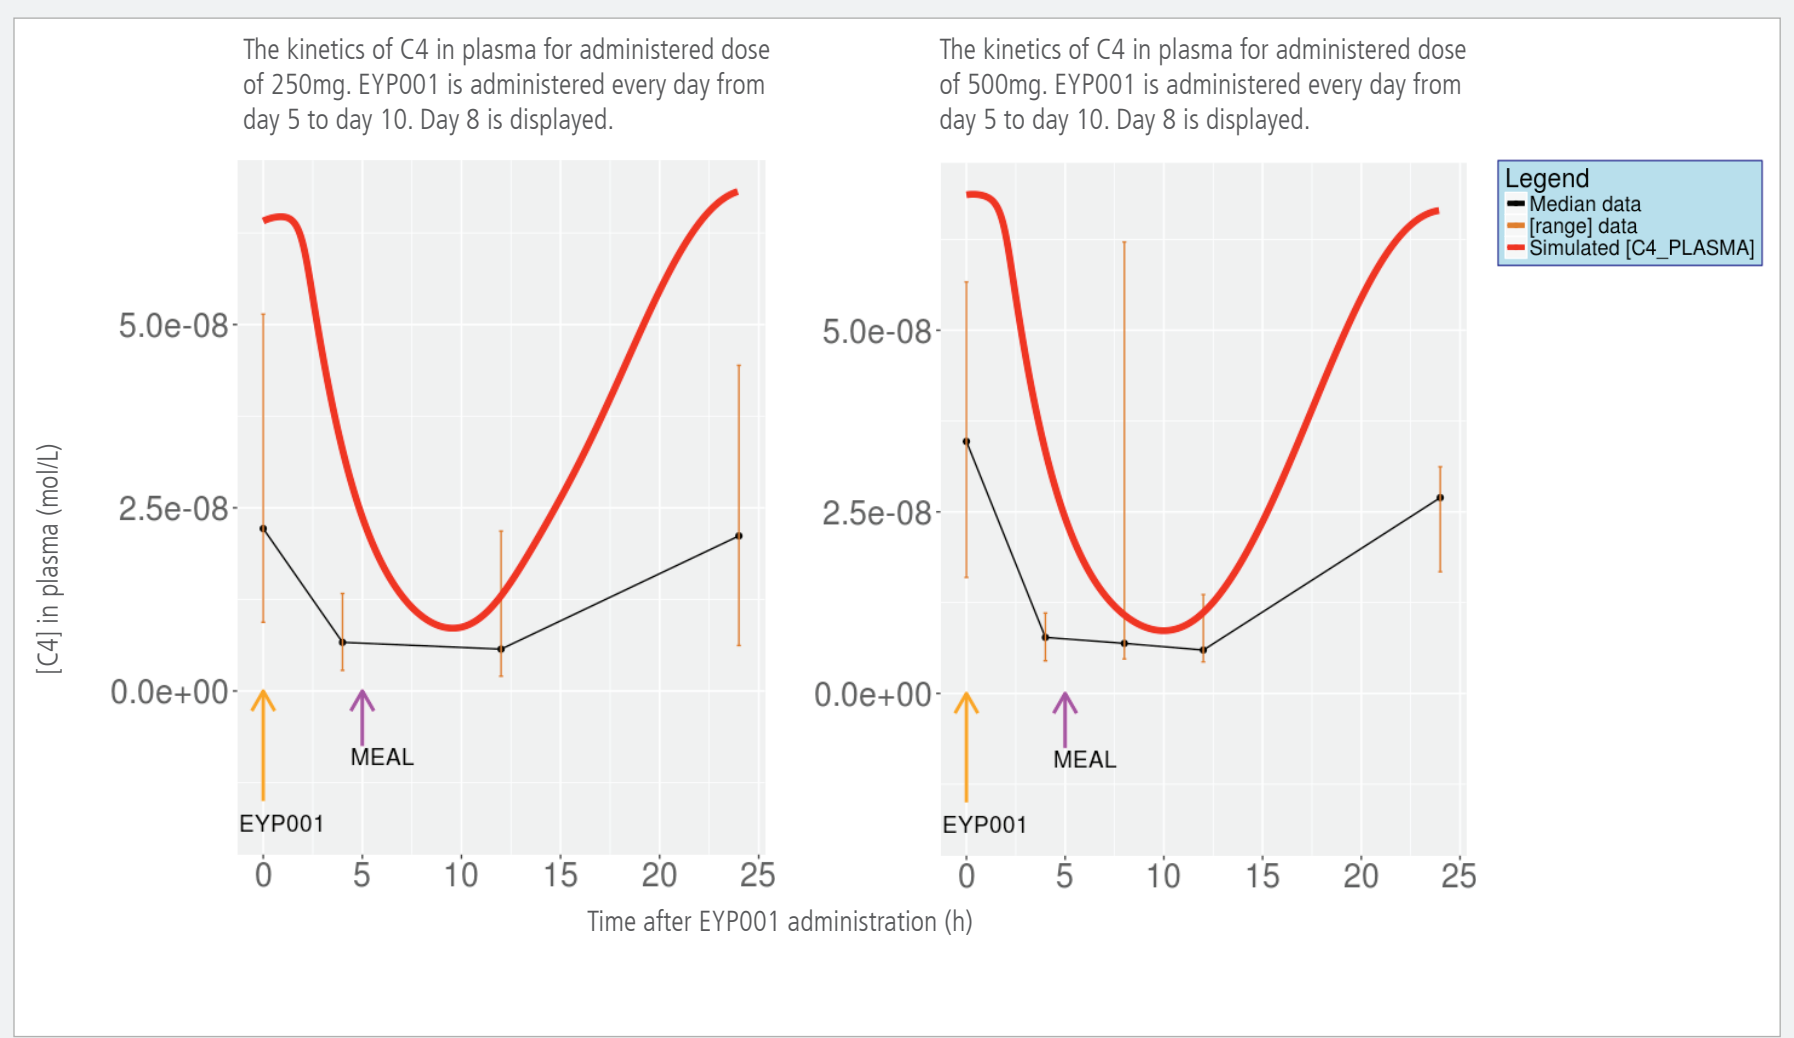
The kinetics of C4 in plasma for administered dose of 250mg. EYP001 is administered every day from day 5 to day 10. Day 8 is displayed.
The kinetics of C4 in plasma for administered dose of 500mg. EYP001 is administered every day from day 5 to day 10. Day 8 is displayed.
[C4] in plasma (mol/L)
Time after EYP001 administration (h)
EYP001
MEAL
Legend
Median data
[range] data
Simulated [C4_PLASMA]

Figure 3a: kinetics of C4 biomarkers in plasma for the administered doses of EYP001 of 250mg (on the left) and 500mg (on the right). 30mg, 60mg, 120mg and 800mg doses were also successfully reproduced (not presented). Black dots separated by black lines and brown bars represent respectively the median data and the minimum-maximum range of experimental data. Red solid lines represent the simulated concentration of biomarkers.

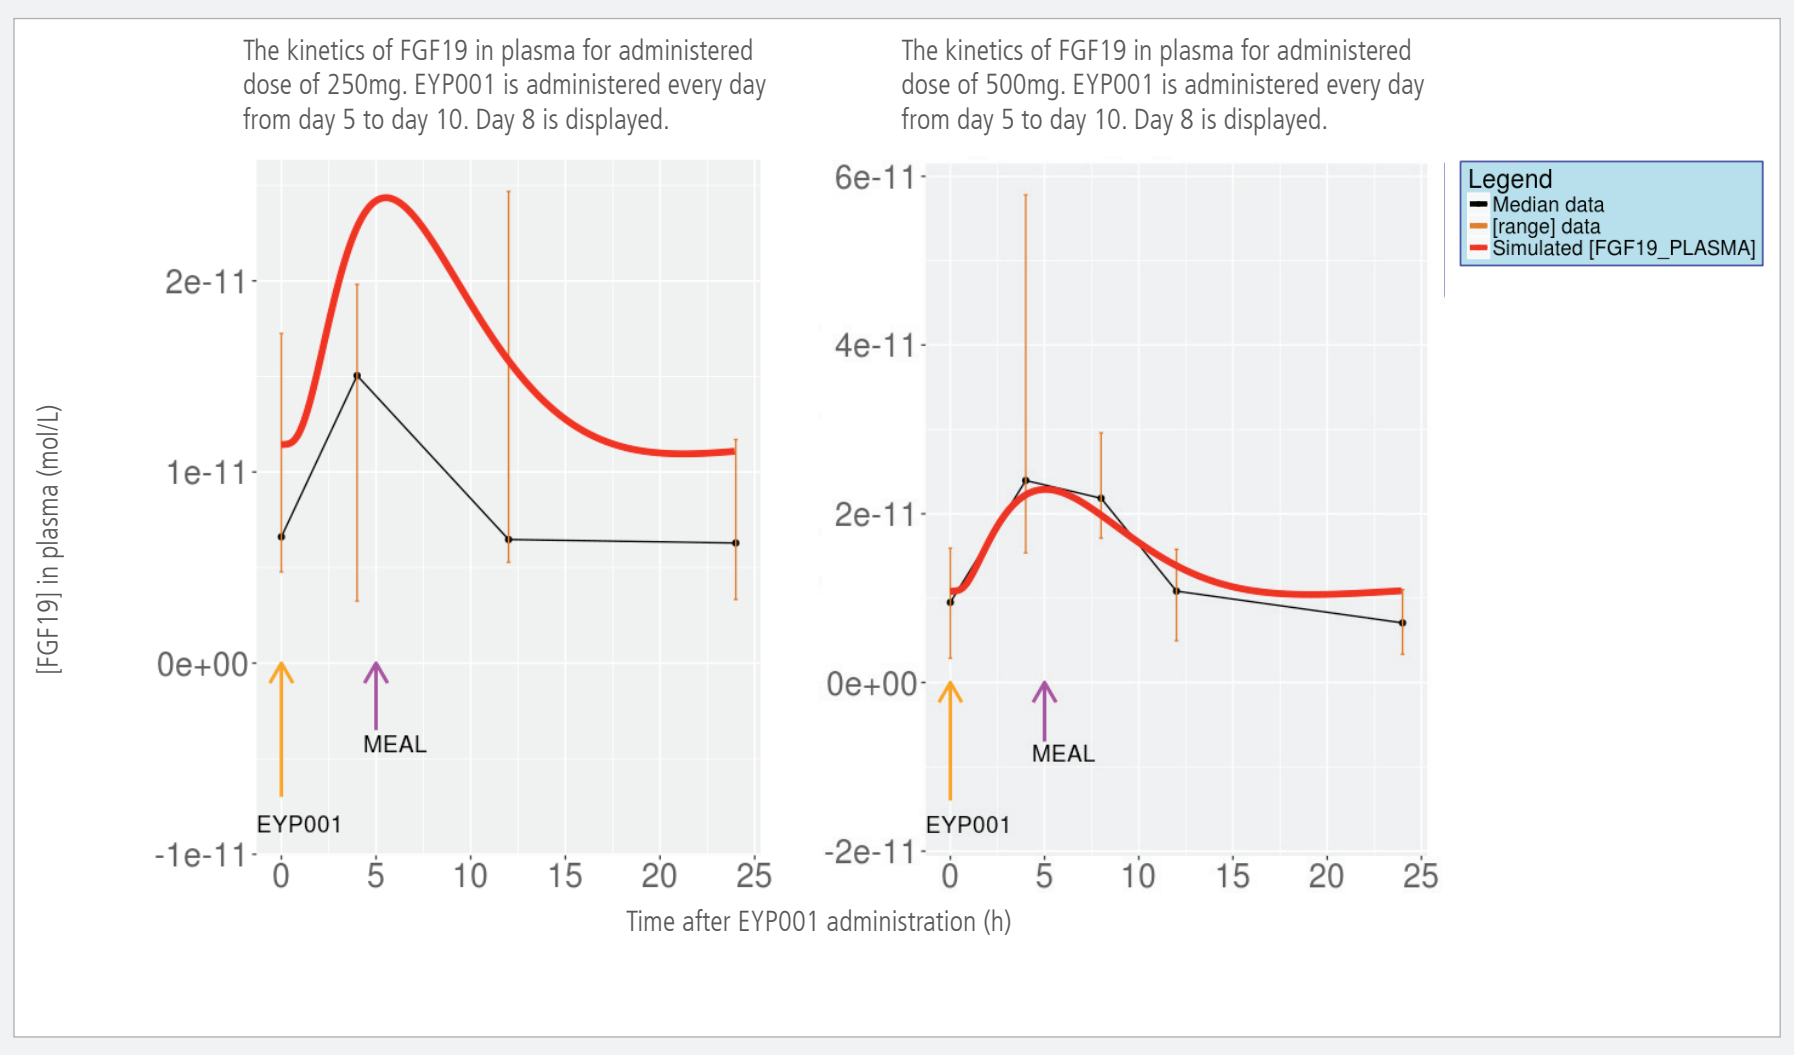
The kinetics of FGF19 in plasma for administered dose of 250mg. EYP001 is administered every day from day 5 to day 10. Day 8 is displayed.
The kinetics of FGF19 in plasma for administered dose of 500mg. EYP001 is administered every day from day 5 to day 10. Day 8 is displayed.
[FGF19] in plasma (mol/L)
Time after EYP001 administration (h)
EYP001
MEAL
Legend
Median data
[range] data
Simulated [FGF19_PLASMA]

Figure 3b: kinetics of FGF19 biomarkers in plasma for the administered doses of EYP001 of 250mg (on the left) and 500mg (on the right). 30mg, 60mg, 120mg and 800mg doses were also successfully reproduced (not presented). Black dots separated by black lines and brown bars represent respectively the median data and the minimum-maximum range of experimental data. Red solid lines represent the simulated concentration of biomarkers.

in silico HBV DNA outputs predictions indicated that prolonged, but not short lasting, FXR agonism with EYP001a inhibited viral replication (Fig. 4).

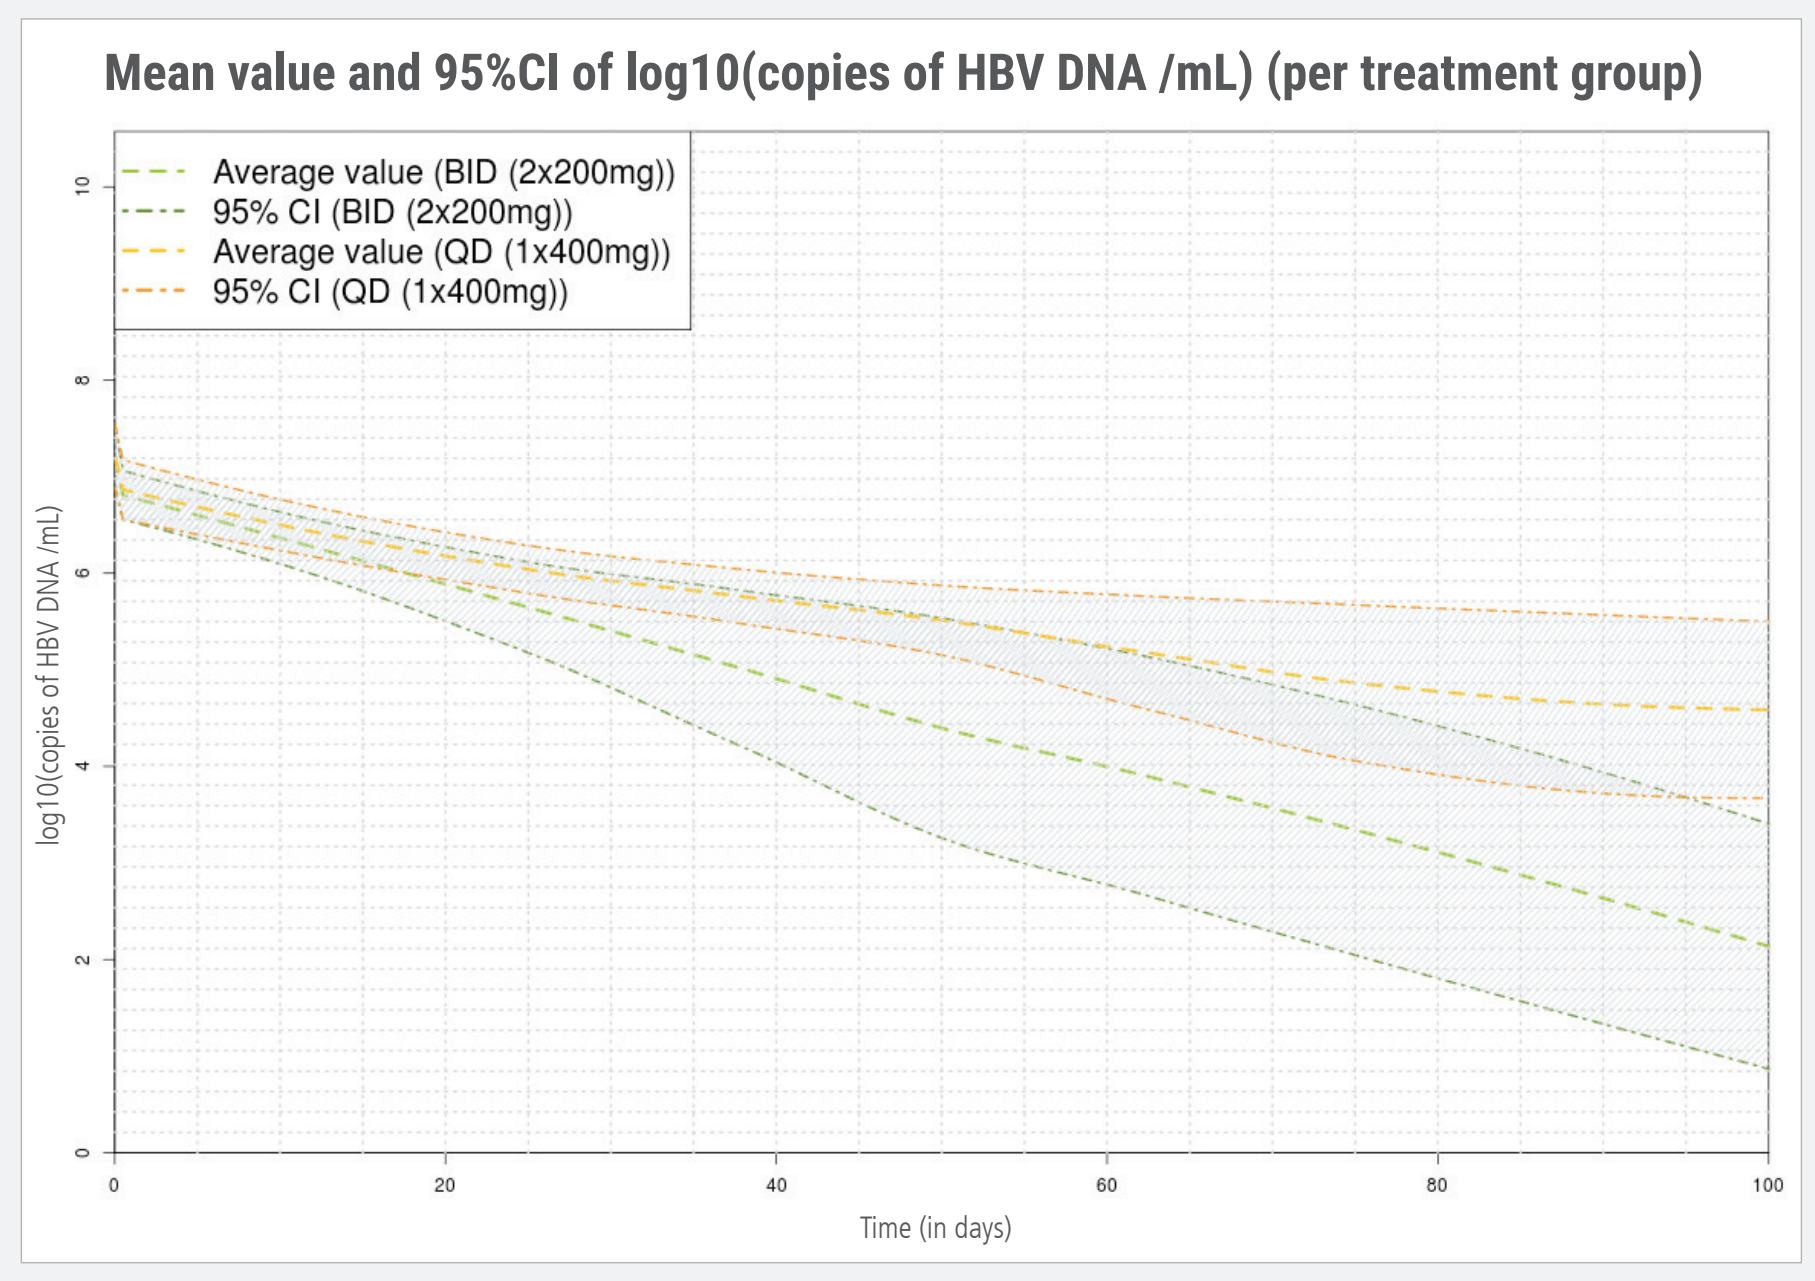
Mean value and 95%CI of log10(copies of HBV DNA /mL) (per treatment group)
Average value (BID (2x200mg))
95% CI (BID (2x200mg))
Average value (QD (1x400mg))
95% CI (QD (1x400mg))
log10(copies of HBV DNA /mL)
Time (in days)

Figure 4: prediction of the evolution of hepatitis B viral DNA (log10 of copies per mL) over 100 days of treatment. The two EYP001a treatment regimens (BID 200mg in green, QD 400mg in orange) are administered at t=0.

Various combinations of dosing regimens with associated cholesterol dietary intakes were tested (Fig. 5) and it was established that 200mg EYP001a BID dosing was an appropriate efficacious regimen (Fig. 6).

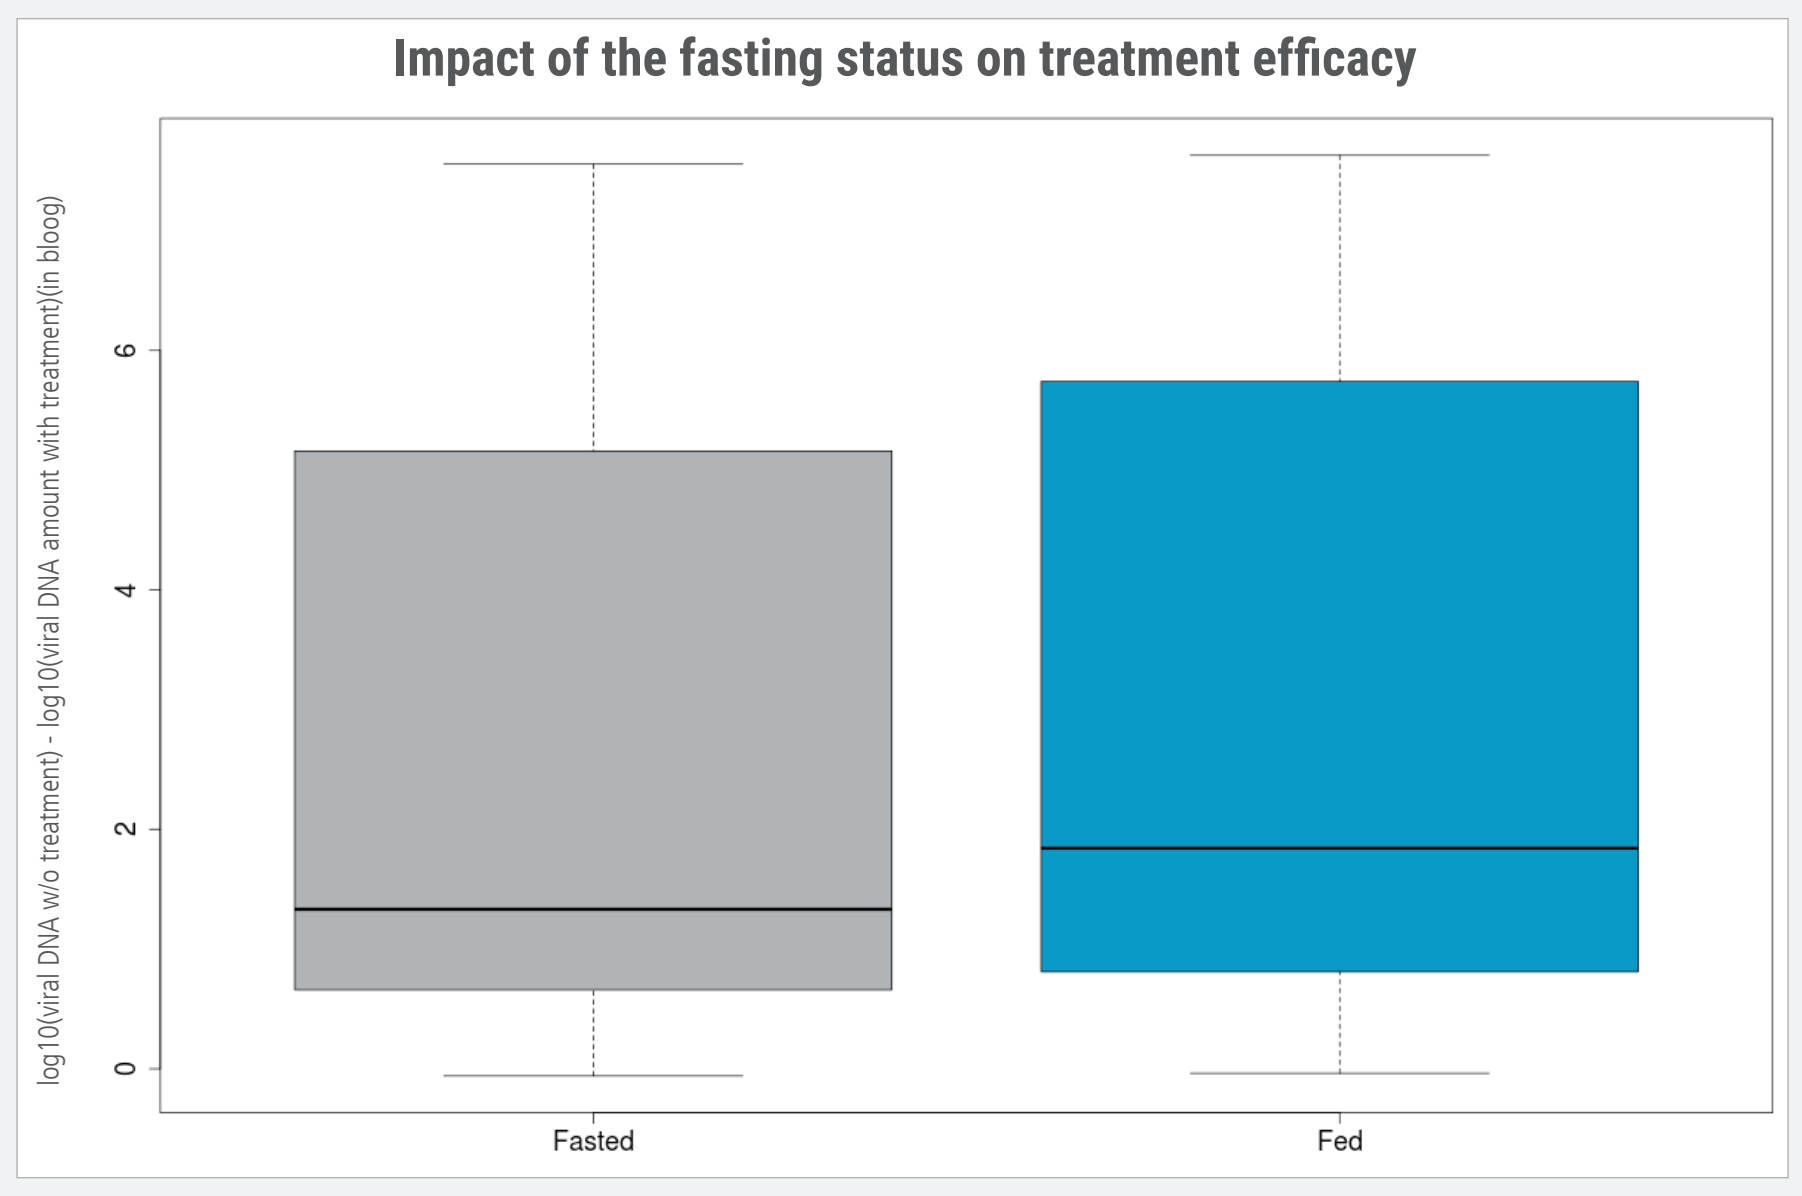
Impact of the fasting status on treatment efficacy
log10(viral DNA w/o treatment) - log10(viral DNA amount with treatment)(in bloog)
Fasted
Fed

Figure 5: simulation on 1,000 virtual patients of the impact of fasting prior to the drug administration on its efficacy. No major differences can be seen between the two statuses with in grey the fasted group and in blue the fed group prior EYP001a administration.

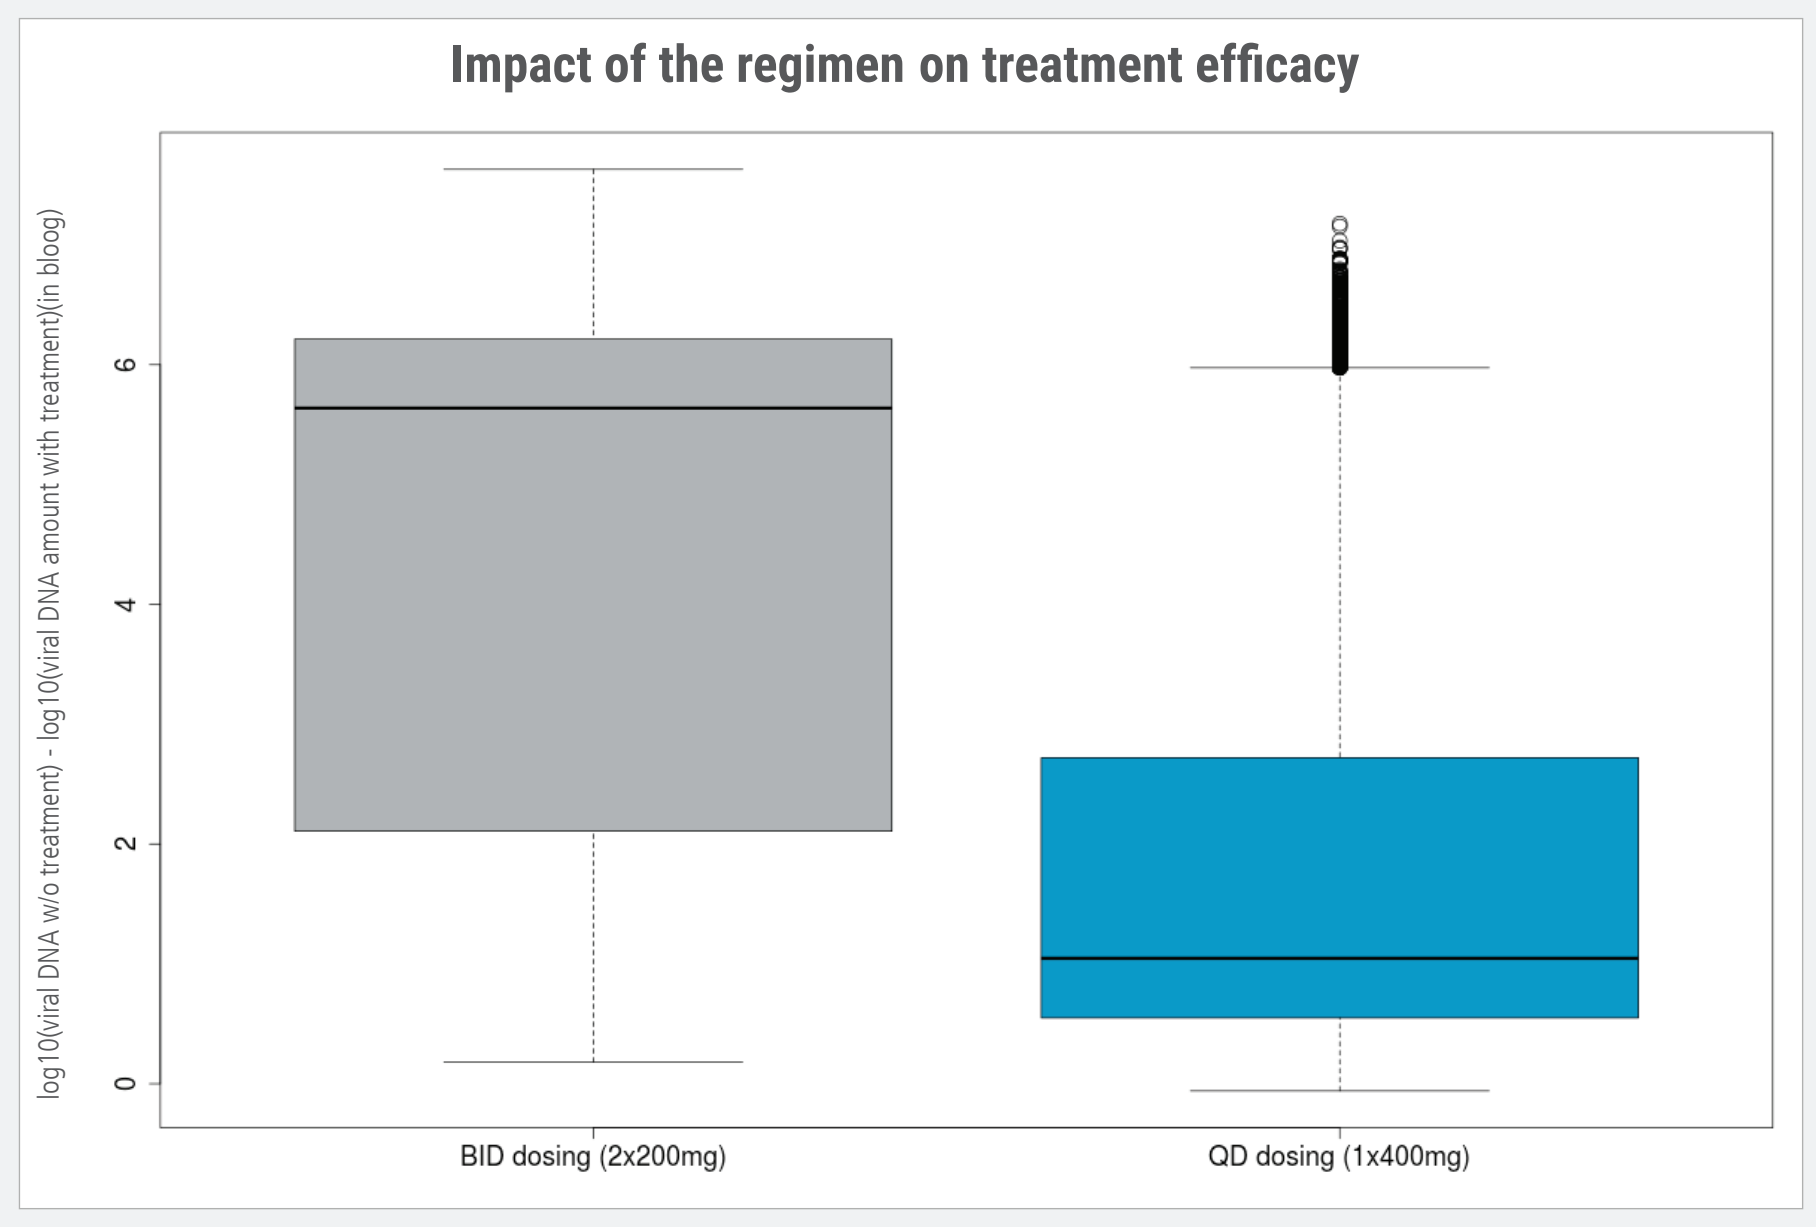
Impact of the regimen on treatment efficacy
log10(viral DNA w/o treatment) - log10(viral DNA amount with treatment)(in bloog)
BID dosing (2x200mg)
QD dosing (1x400mg)

Figure 6: comparison of the observed treatment efficacy (simulation on 1,000 virtual patients) between the drug regimens. The BID 200mg dosing (blue boxplot) appears to be more efficient than the QD 400mg (grey boxplot).

DISCUSSION & CONCLUSIONS

/ The *in silico* model reproduced well EYP001a plasma concentrations as well as the dynamics of C4 and FGF19 in blood.

/ The *in silico* model predicted the viral response in a virtual HBV infected population (1,000 virtual patients).

/ The strong predictability of our simulation approach using *in silico* modeling could be used to determine an a priori better dosing regimen in chronic HBV patients. We predicted a superior efficacy of the treatment when it is administered in two separate doses (BID 200mg) instead of only one full dose (QD 400mg). The average reduction of the number of viral particles tends to be higher among virtual patients who received two doses of 200mg.

/ The differences in terms of efficacy between fasted and fed before the administration of the treatment appear to be minimal. However, it is important to note that we did not integrate a complete and mechanistic model of nutrition (cholesterol dietary intakes only).

/ Once quantitatively validated with data from both in vitro HepaRG experiments and *in vivo* HBV infected subjects, this *in silico* model will be used to explore other FXR agonist treatment strategies and to identify best responders in the population to be tested in coming phase II HBV trials.

ACKNOWLEDGEMENTS

This study was supported by Enyo Pharma SA. We want to thank Calvagone team for the exchange of the PKPD data.

REFERENCES

[1] Pras Pathmanathan, Richard A. Gray, Vicente J. Romero, Tina M. Morrison, 2017, 'Applicability Analysis of Validation Evidence for Biomedical Computational Models', Journal of Verification, Validation and Uncertainty Quantification, vol. 2, no. 2, pp. (doi:10.1115/1.4037671).

CM v1.1 2018-05-23

NOVADISCOVERY, Place Giovanni da Verrazzano, 69009 Lyon, France, contact@novadiscovery.com
ENYO PHARMA, 321 avenue Jean Jaurès, 69007 Lyon, France, contact@enyopharma.com

www.novadiscovery.com