Interplay and Recruitment of FXR and HBx on cccDNA and Role on Viral Transcription

B. LACOMBE [1,2,3], C. MENARD [1,2,3], P-O. VIDALAIN [1,2,3], Y. JACOB [6], R. DARTEIL [5], J. VONDERSCHER [5], V. LOTTEAU [1,2,3], C. RAMIERE [1,2,3,4] and P. ANDRE [1,2,3,4]

1 Centre International de Recherche en Infectiologie, Institut National de la Sante et de la Recherche Médicale Unité 1111, Centre National de la Recherche Scientifique Unités Mixte de Recherche 5308, Lyon, France.
2 Ecole Normale Supérieure de Lyon, Lyon, France . 3 Université Claude Bernard Lyon 1, Villeurbanne, France. 4 Laboratoire de Virologie, Hôpital de la Croix–Rousse, Hospices Civils de Lyon, Lyon, France. 5 Enyo Pharma, Lyon, France. 6 Département de Virologie, Unité de Génétique Moléculaire des Virus à ARN (GMVR), Institut Pasteur, Centre National de la Recherche Scientifique Unités Mixte de Recherche 3569, Université Paris Diderot, Sorbonne Paris Cité, Paris, France.

CONTEXT

Entry bile acids (BA) receptor, NTCP, is the first line of evidence that their is a link between BA metabolism and Hepatitits B virus (HBV) replication in hepatocytes. Then, viral replication modulates gene expression that are involved in BA matabolism and especially Farnesoid X (FXR) nuclear receptor. This nuclear receptor actively contributes to BA homeostasis in liver and intestine. Finally, FXR ligands modulate its activity and protein expression. They affect completion of HBV minichromosome but also viral replication in hepatocytes. Eliminate cccDNA or silence viral transcription is the first step to a functional cure.

OBJECTIVES

Many evidences show that bile acids (BA) nuclear receptor, FXR, is a proviral factor for HBV replication. It directly links on FXR response elements located on HBV EN2/Cp promoter (1). Its ligands have a demonstrated antiviral activity against HBV replication (2). It has also been proved that FXR is important for cccDNA completion and transcription of newly viral matrix (3). In addition, it has been suggested that murine FXR interacts with the viral transcriptional regulator HBx (4). We further aim to decipher the relationship and the interaction between FXR and HBx to better understand how they can both interfere with viral transcription.

MATERIELS & METHODS

Cells:
- 293T (human embryonic kidney cell line)
- Huh7 (human hepatocarcinoma cell line)
- Differentiated HepaRG (dHepaRG)
- Viral infection: HBV WT (from HepaD38), HBV ΔHBx (from HepG2

Experiments:
- Chromatin immunoprecipitation (ChIP)
- qPCR
- Western Blot
- Nanoluc two hybrid (N2H) (5)

REFERENCES

(1) **Ramiere C. et al.** Transactivation of the Hepatitis B Virus Core Promoter by the Nuclear Receptor FXRα. *J Virol 2008; 82;: 10832-10840*.

(2) **Radreau P. et al.** Reciprocal regulation of farnesoid X receptor α activity and hepatitis B virus replication in differentiated HepaRG cells and primary human hepatocytes. *FASEB J 2016; 30;: 3146-3154*.

(3) **Mouzannar K. et al.** Farnesoid X receptor α is a proviral host factor for hepatitis B virus that is inhibited by ligands in vitro and in vivo. *FASEB J 2018; 33;: 2472-2483*.

(4) **Niu Y. et al.** Farnesoid X receptor ablation sensitizes mice to hepatitis B virus X protein-induced hepatocarcinogenesis. *Hepatology 2017; 65;: 893-906*.

(5) **Choi G.S. et al.** Maximizing binary interactome mapping with a minimal number of assays. *Nat communications 2019; 10; 3907*.

ACKNOWLEDGEMENTS

This work is supported by ANRS.
B. Lacombe and C. Ménard are supported by a collaborative project with Enyo Pharma, Lyon, France.
V. Lotteau and P. Andre are consultants for Enyo Pharme

CONTACT

patrice.andre@inserm.fr

RESULTS

Figure 1: FXR interacts with HBx and the AF1 domain is not necessary for this interaction. We took advantages of the Nanoluc two hybrid assay (5) to investigate the interaction between FXR and HBx. NLR values are calculated with relative Luciferase units detected from FXR (full-length and several domains) and HBx compared to Luciferase units of related controls. Interaction is positive if NLR >1.

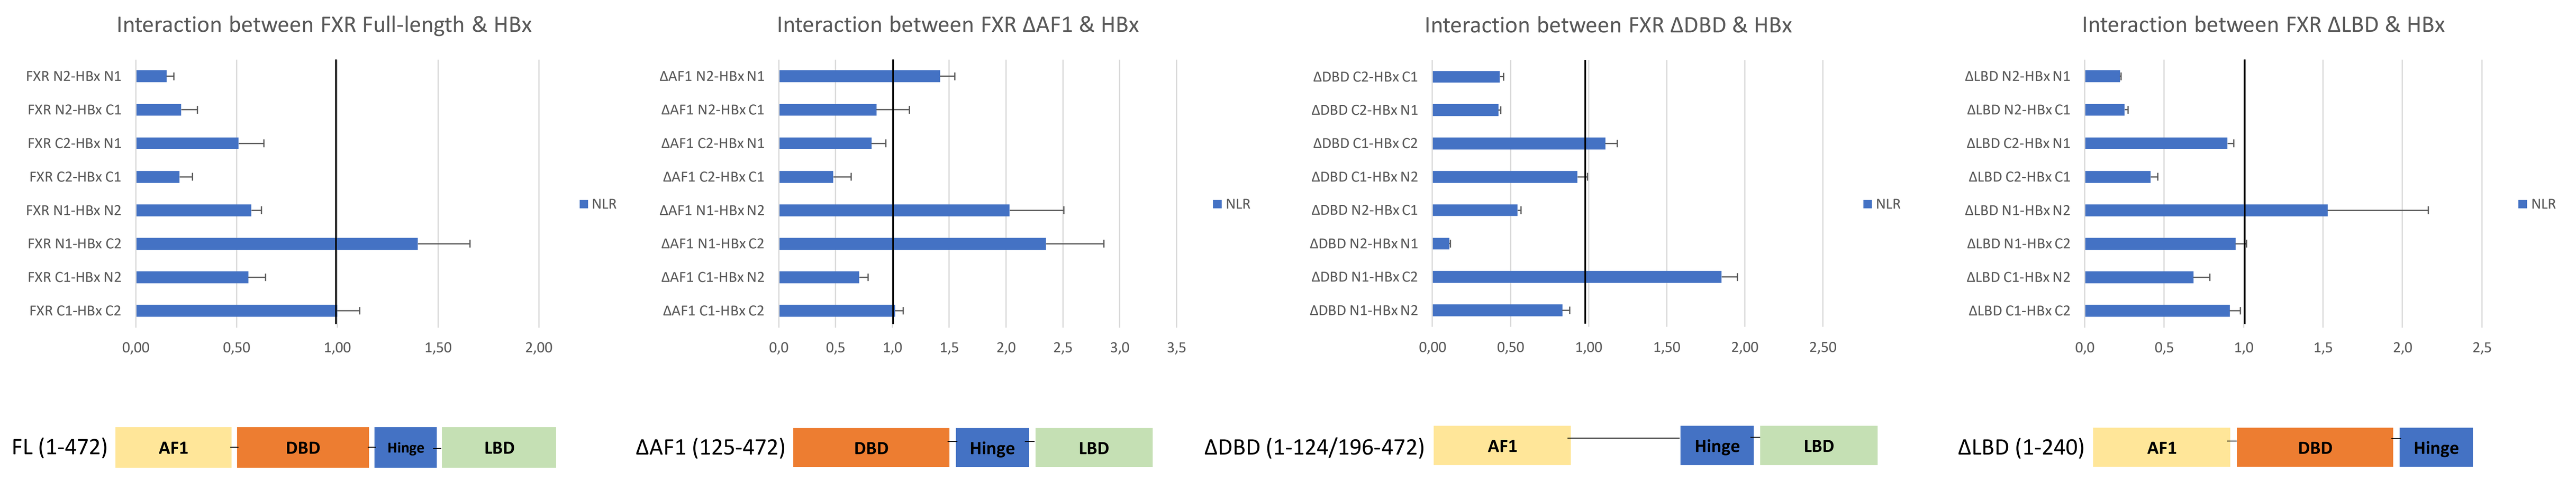

Interaction between FXR Full-length & HBx
Interaction between FXR ΔAF1 & HBx
Interaction between FXR ΔDBD & HBx
Interaction between FXR ΔLBD & HBx
NLR
FL (1-472)
AF1
DBD
Hinge
LBD
ΔAF1 (125-472)
ΔDBD (1-124/196-472)
ΔLBD (1-240)

Figure 2: In Huh7, GW4064 releases FXR from EN2/Cp promoter but stabilizes it on BSEP promoter. We performed ChIP experiments in Huh7 transfected with EN2/Cp promoter plasmid. Immunoprecipitation were performed with H3K4me3 and FXR antibodies. Regions inside promoters were analyzed by qPCR.

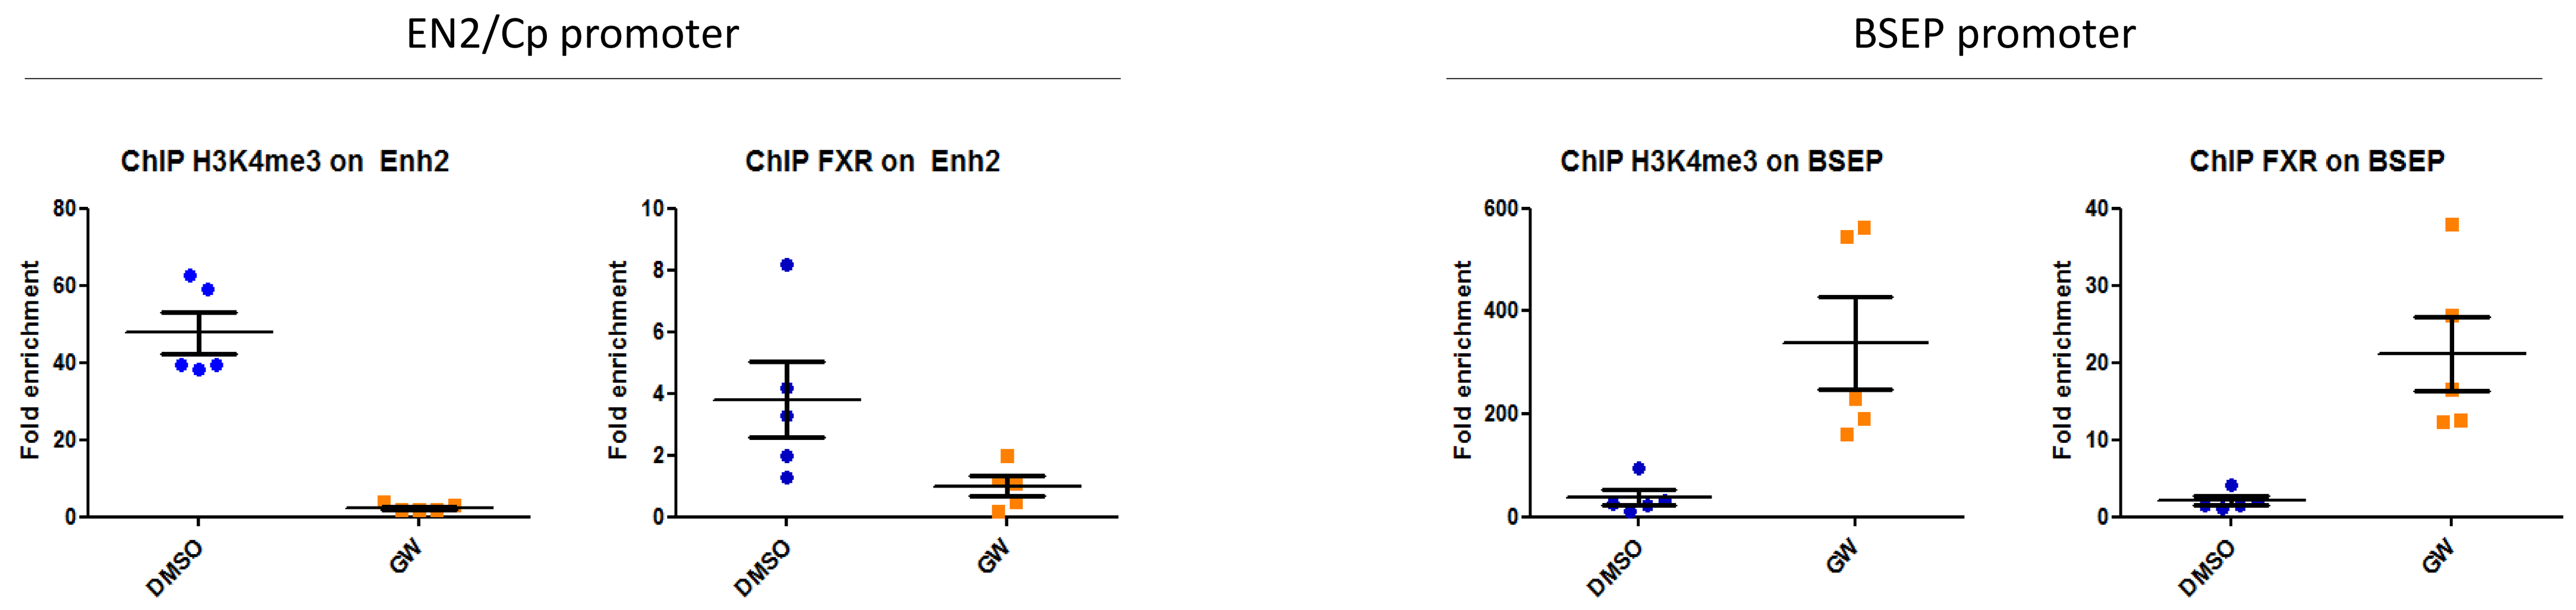

EN2/Cp promoter
BSEP promoter
ChIP H3K4me3 on Enh2
ChIP FXR on Enh2
ChIP H3K4me3 on BSEP
ChIP FXR on BSEP
Fold enrichment
DMSO
GW

Figure 3: In relevant hepatocytes model, dHepaRG, GW4064 decreases FXR at protein and mRNA level. It as for consequences to diminish viral transcription in HBV WT infected cells. When dHepaRG were treated with GW4064 during 72h, FXR is downregulated at protein and mRNA level. Viral transcription is decreased but BSEP is still strongly upregulated.

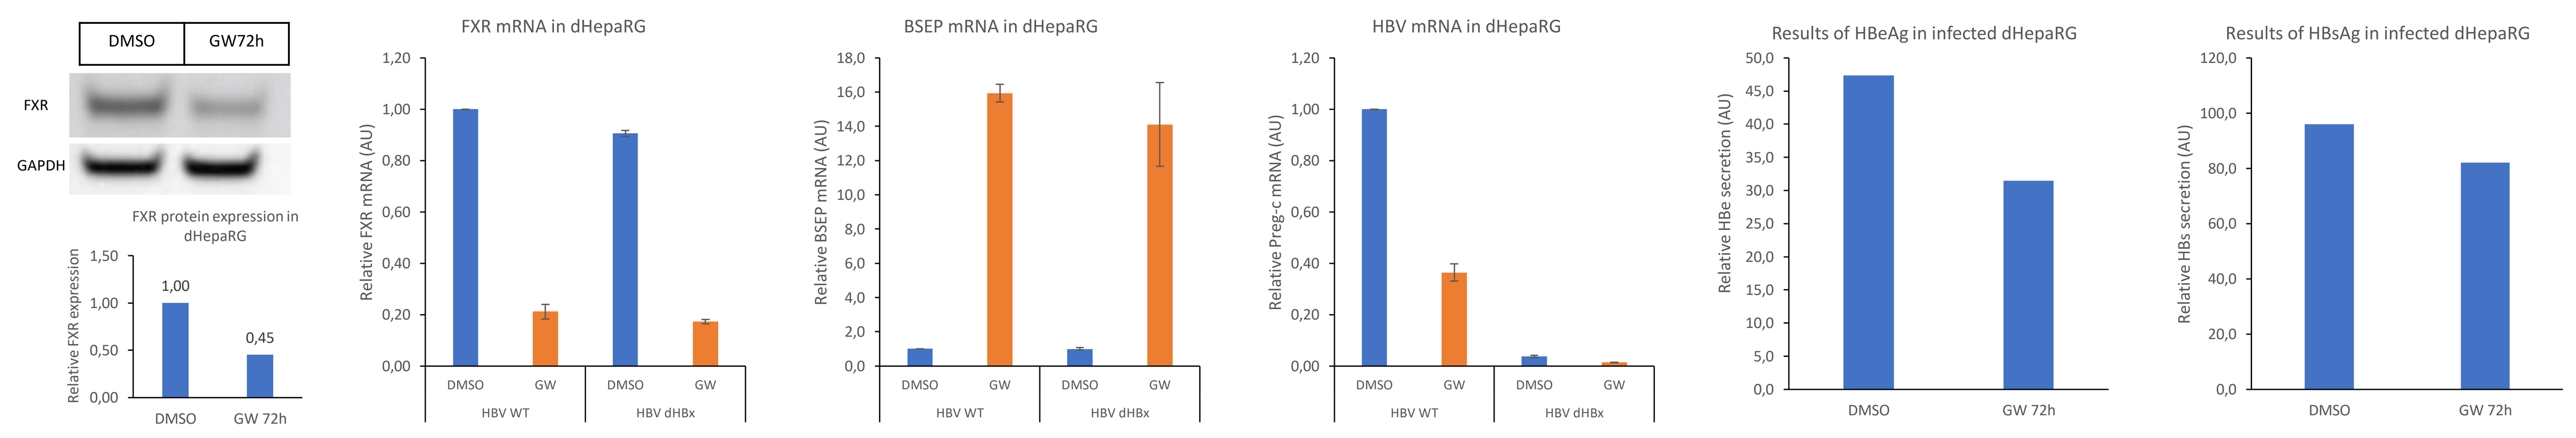

DMSO
GW72h
FXR
GAPDH
FXR protein expression in dHepaRG
Relative FXR expression
FXR mRNA in dHepaRG
Relative FXR mRNA (AU)
BSEP mRNA in dHepaRG
Relative BSEP mRNA (AU)
HBV mRNA in dHepaRG
Relative Preg-c mRNA (AU)
HBV WT
HBV dHBx
Results of HBeAg in infected dHepaRG
Relative HBe secretion (AU)
Results of HBsAg in infected dHepaRG
Relative HBs secretion (AU)
GW 72h

Figure 4: GW4064 decreases RNA Polymerase II recruitment at EN2/Cp promoter but increases its recruitment on BSEP promoter. We performed ChIP experiments in dHepaRG infected with HBV WT. Immunoprecipitation were performed with several histones marks and polymerase forms antibodies. Regions inside promoters were analyzed by qPCR.

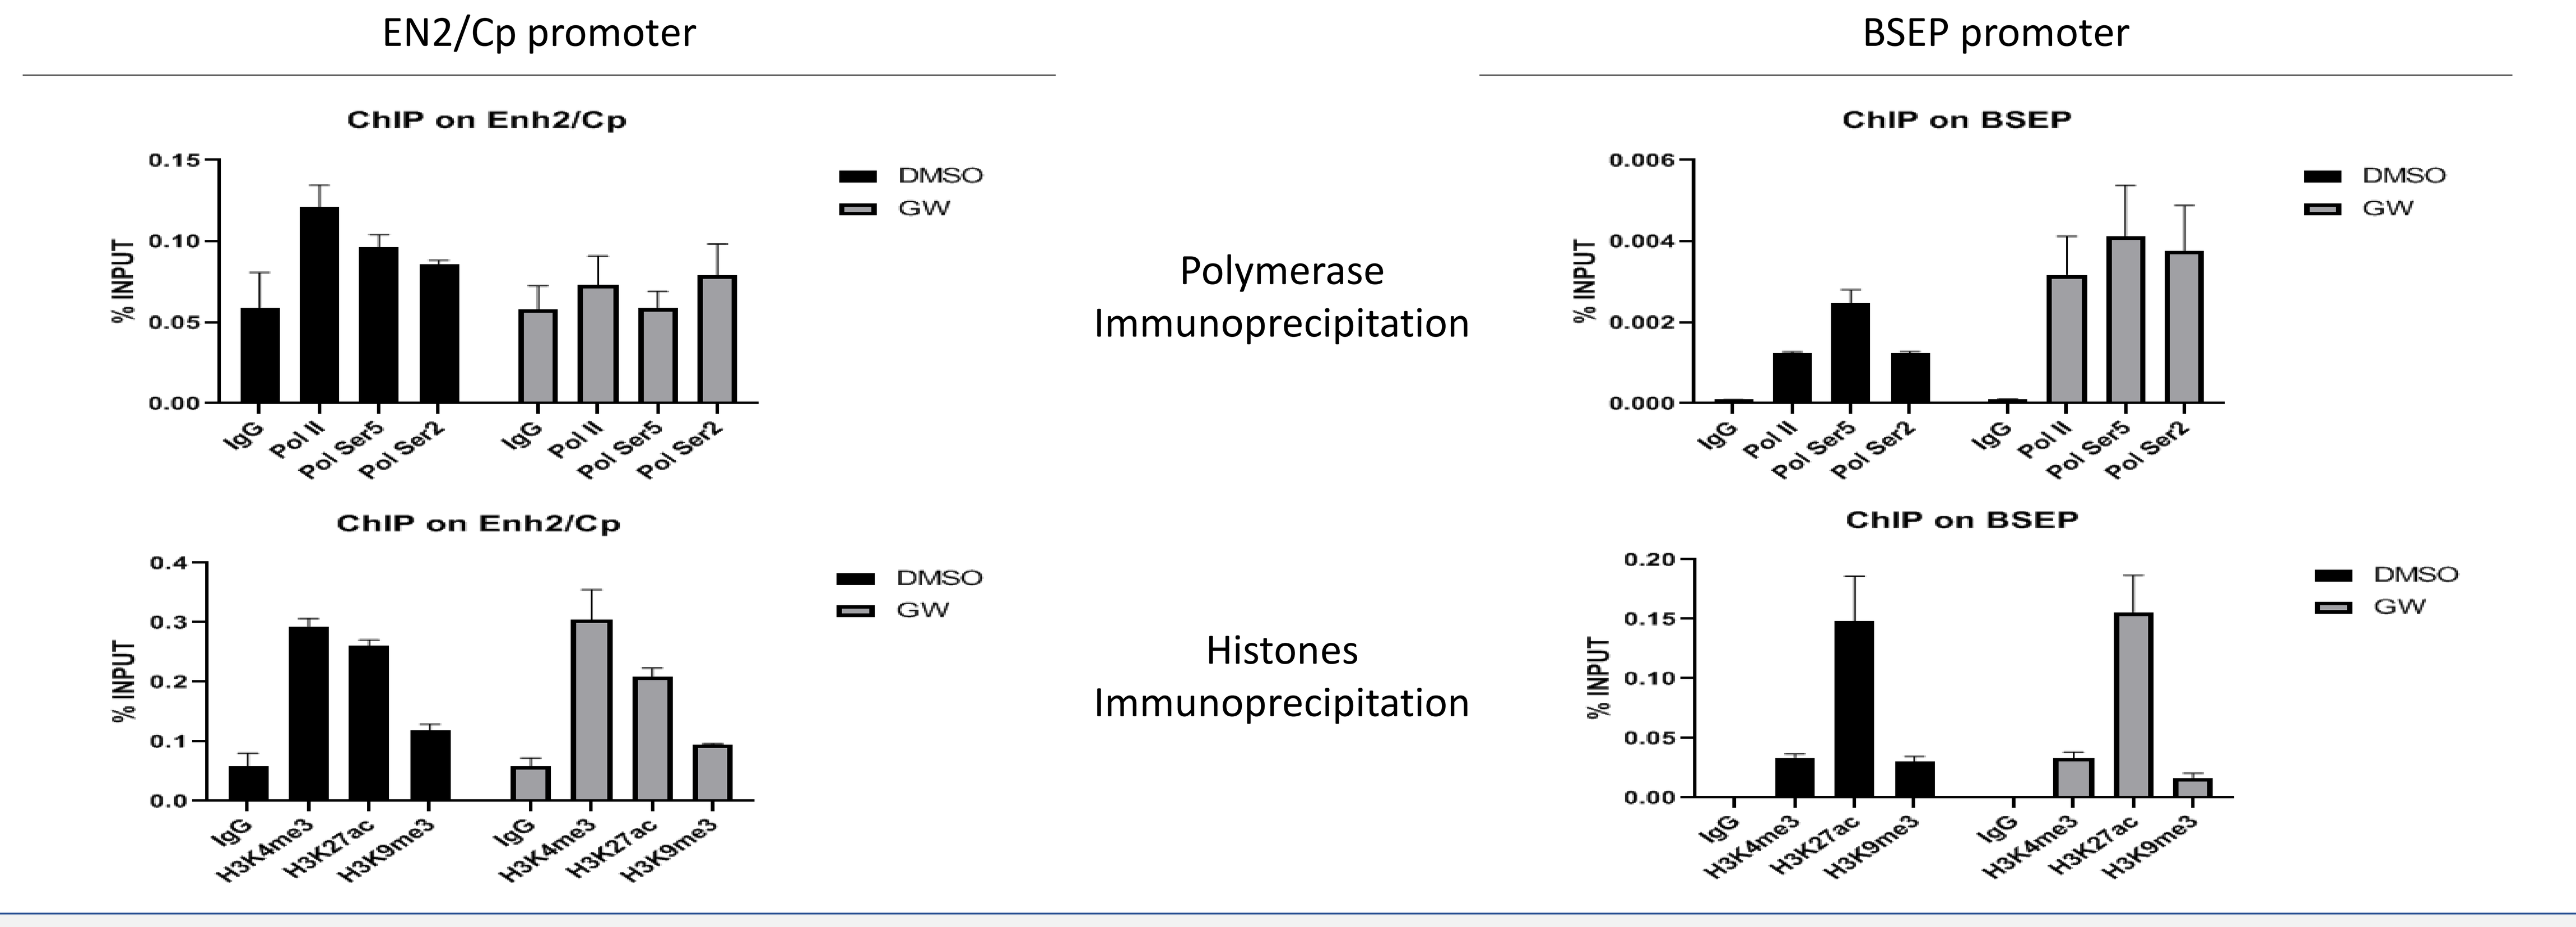

EN2/Cp promoter
BSEP promoter
ChIP on Enh2/Cp
ChIP on BSEP
% INPUT
DMSO
GW
IgG
Pol II
Pol Ser5
Pol Ser2
H3K4me3
H3K27ac
H3K9me3
Polymerase Immunoprecipitation
Histones Immunoprecipitation

CONCLUSIONS

The data suggest that:

- proviral activity of FXR depends on the fixation of FXR on EN2/Cp promoter. Also on the fact that FXR interacts with HBx and by the way could bring HBx near the EN2/Cp promoter to regulate viral transcription.

- antiviral activity of GW4064 relies on three actions from the ligand. The first is the downregulation of FXR protein and mRNA level in hepatocytes. The second is the releasing of FXR from EN2/Cp. Degrade or release FXR from the viral promoter could get away HBx from this regulated area and by consequence blocks HBx action. The last is the decrease of RNA Pol II recruitment on EN2/Cp promoter.

GW4064 is a strong HBV transcriptional inhibitor.