POPULATION PK-PD MODELLING OF C4 AND FGF19 CONCENTRATIONS AFTER ADMINISTRATION OF THE FXR AGONIST EYP001 IN HEALTHY AND HBV INFECTED SUBJECTS

Pierrillas PB[1], Scalfaro P[2], André P[3], Darteil R[2], Barzic N[2], Couchoux H[2], Vonderscher J[2] and Laveille C[1]

[1] Calvagone, Liergues, France ; [2] ENYO Pharma SA, Lyon, France ; [3] Centre International de Recherche en Infectiologie (CIRI) - INSERM U1111 – CNRS UMR5308 - Université Lyon 1- ENS de Lyon

INTRODUCTION

EYP001a is a non bile salt, second generation agonist of the nuclear farnesoid X receptor (FXR) which binds bile acids. FXR agonists, originally discovered for a therapy of non-alcoholic steato-hepatitis, primary biliary cholangitis and metabolic syndrome, were found to have anti-viral activity on Hepatitis B virus (HBV) [1].

The objective of this work was to develop a population Pharmacokinetic-Pharmacodynamic model (PK-PD) using nonlinear mixed effect modelling to assess the effect of EYP001a on bile acids pathway in both healthy volunteers and HBV infected patients

PATIENTS AND METHODS

Data/study description (**Table 1**)

Data from four phase 1 studies (including a 4-arm cross-over study to evaluate the impact of food intake and a potential circadian rhythm and a drug-drug interaction study) conducted in healthy volunteers and HBV-infected patients were included in this analysis (after single and repeated doses).

Overall, 2427 EYP001a concentrations, 2080 C4 observations (7α-hydroxy-4-cholesten-3-one, used as surrogate marker for FXR response) and 2032 FGF19 observations (fibroblast growth factor 19) from 180 individuals were considered in the analysis.

Model development

Population PK-PD models were built using nonlinear mixed effect modelling.

A covariate analysis, using a stepwise approach was performed including the investigation of the potential differences between healthy volunteers and HBV infected patients.

Model development, model diagnostics and model simulations were performed using NONMEM 7.3 software (ICON).

Simulations

Simulations of the PK-PD model were performed to investigate C4 and FGF19 profiles for different dosing regimens.

Table 1: Studies' characteristics

	Study C01	Study 102	Study 103	Study 104
Population	Healthy Volunteers	HBV patients	HBV patients	Healthy Volunteers
Number of individuals	80	11	73	16
Type of study	Single and multiple dose	4-arm cross-over	Multiple dose	Drug - drug interaction
Dose	From 30 to 800 mg or placebo	300 mg	From 100 to 400 mg (BID or QD) or placebo	300 mg or placebo

RESULTS

Model development description

PK model

Plasma PK of EYP001a was best described with:

- 2-compartment disposition model
- 4 transit compartments for the absorption phase
- relative bioavailability decreases after repeated administrations and as dose increases

Covariate analysis suggested:

- lower clearance in HBV patients (~25%)
- clearance decreases as bilirubin level increases
- administration under fed condition decreases the absorption rate
- administration in the evening decreases the absorption rate

FGF19 PK-PD model

FGF19 time-course was modelled using:

- a turn-over model
- a K-PD approach [3] to describe the increase of FGF19 production induced by meal intake [4]
- an effect compartment and a sigmoidal function stimulating FGF19 production for EYP001 drug effect

Covariate analysis suggested:

- EYP001a potency decreases as age increases
- EYP001a potency increases as HDL increases
- FGF19 baseline decreases as bodyweight increases
- FGF19 baseline is lower in Polish individuals

C4 PK-PD model

C4 time-course was modelled using:

- a turn-over model with a cosine function to describe the circadian rhythm of C4 production
- an effect compartment [2] and a sigmoidal function inhibiting C4 production for EYP001a drug effect

Covariate analysis suggested:

- C4 production increases as age increases

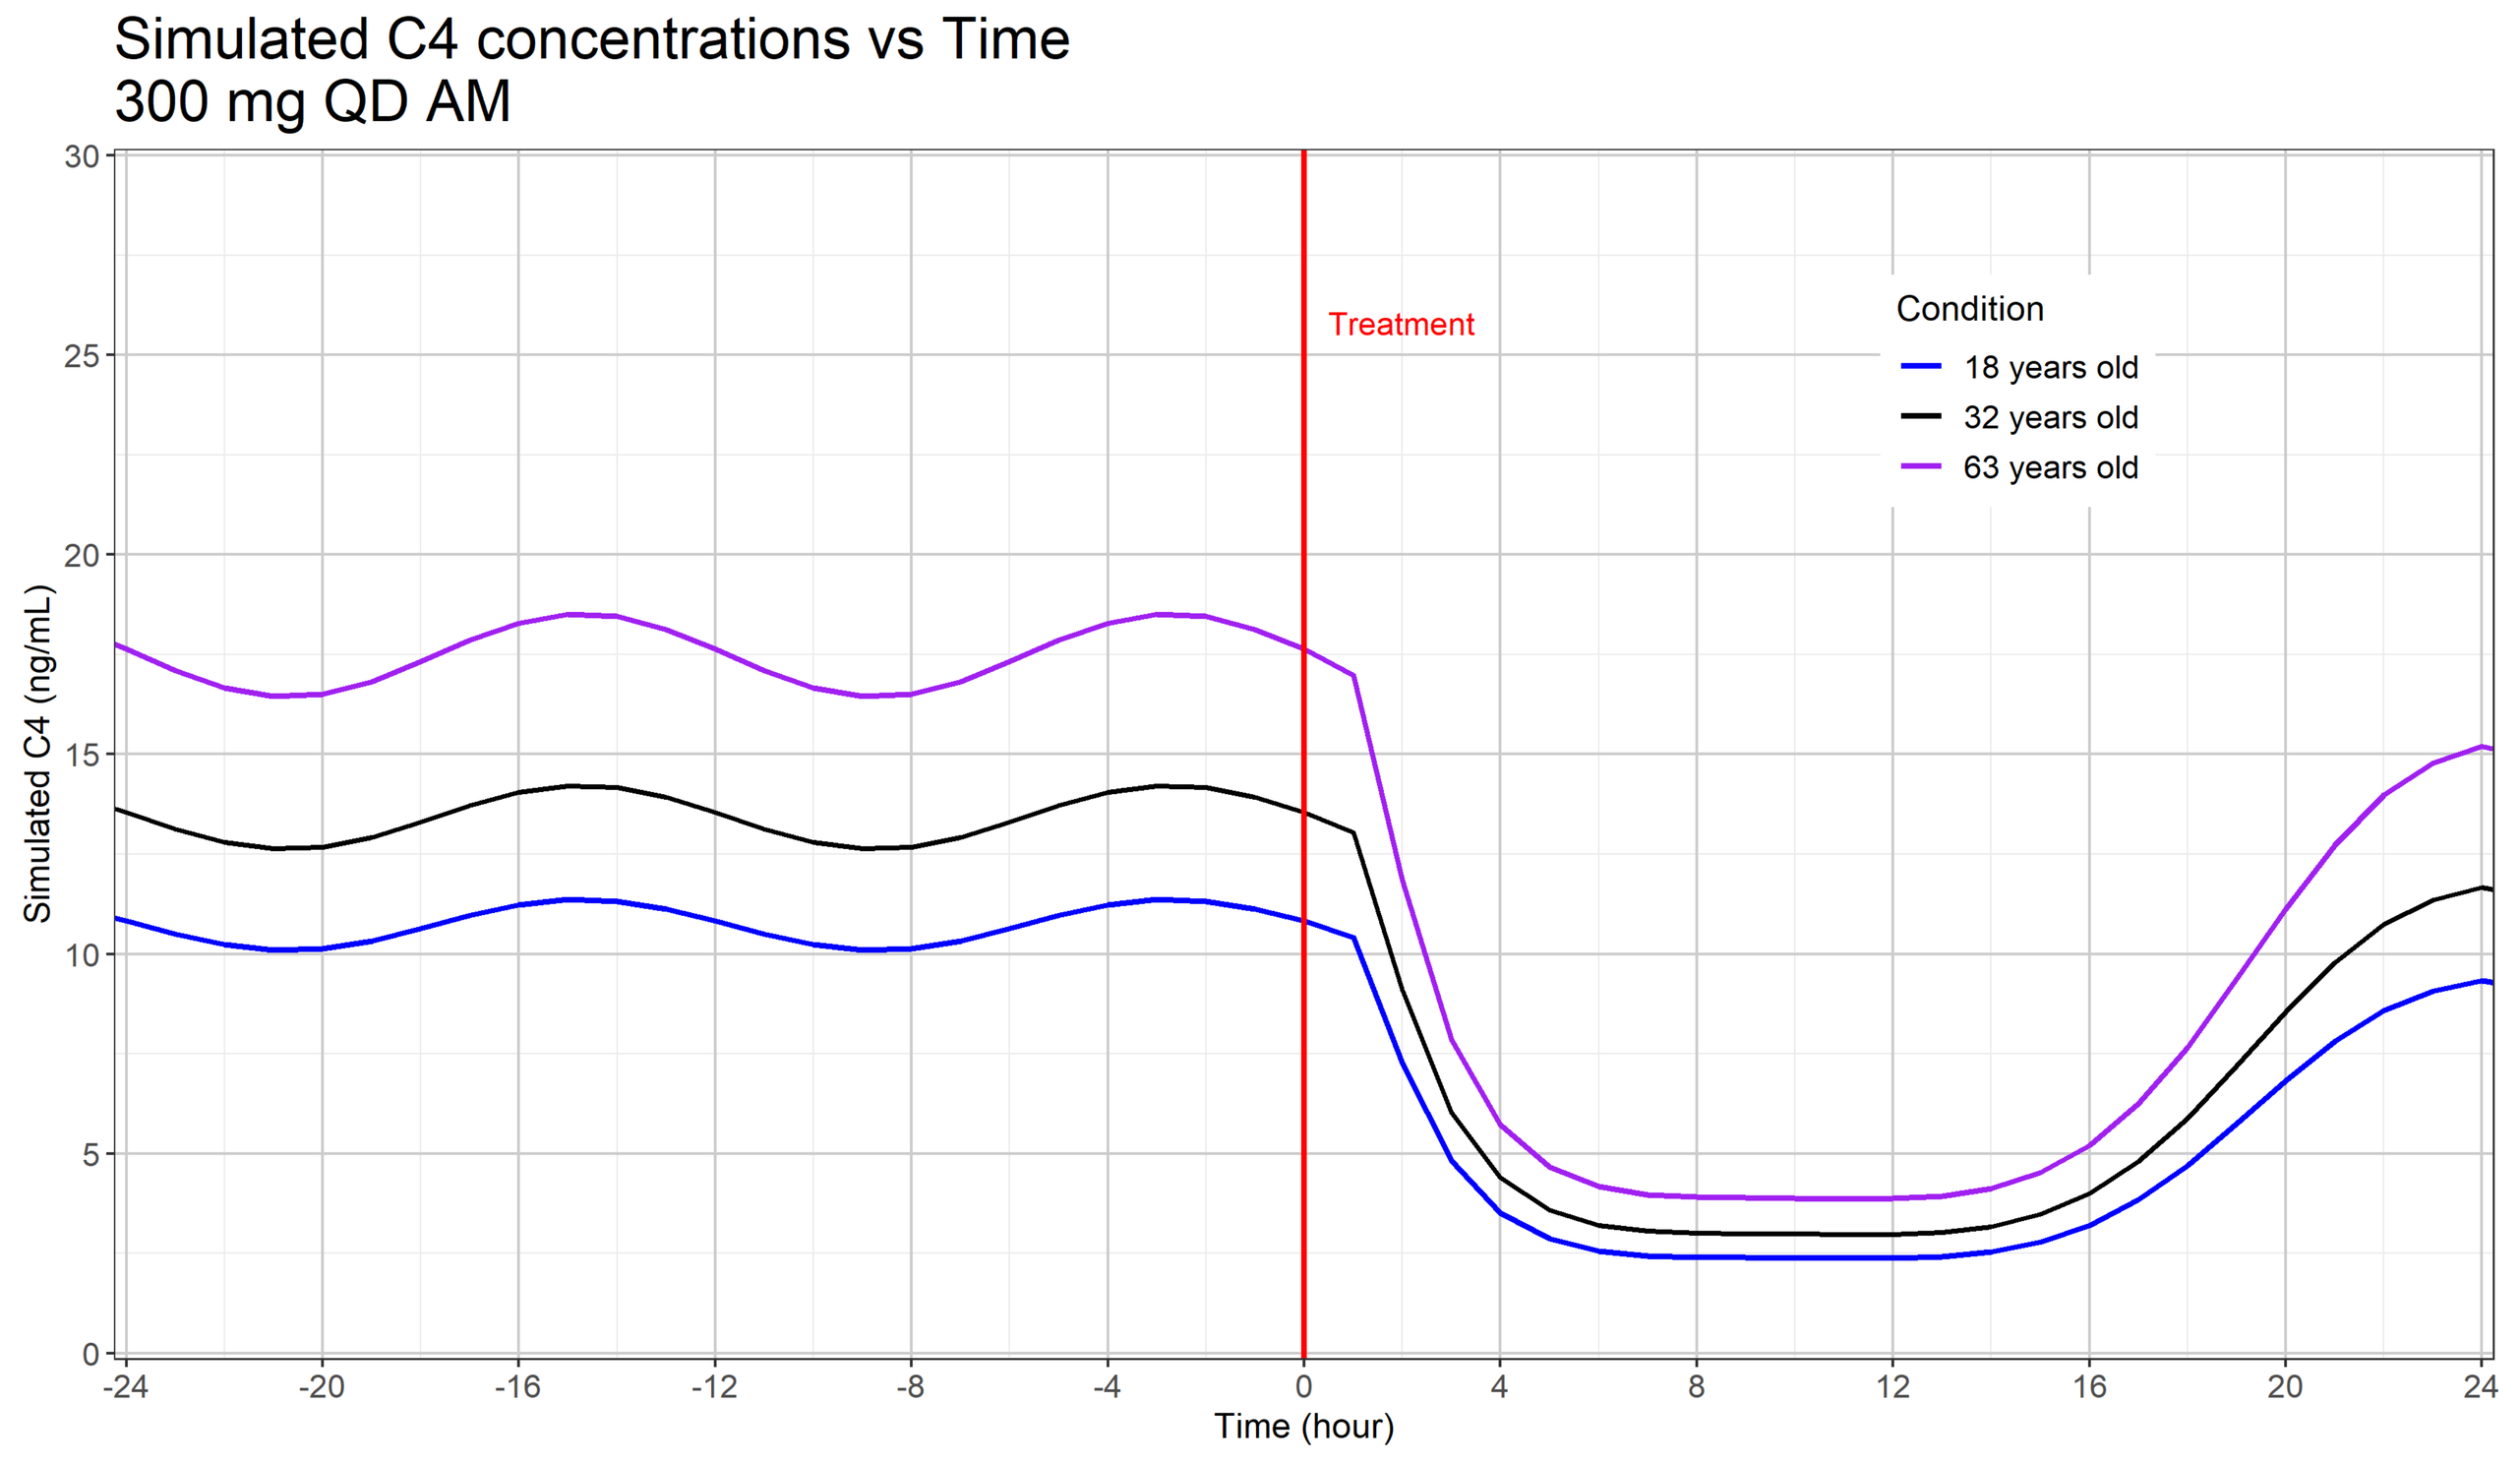
Simulated C4 concentrations vs Time
300 mg QD AM
Treatment
Condition
18 years old
32 years old
63 years old
Simulated C4 (ng/mL)
Time (hour)

Figure 1: Impact of age on C4 kinetic

Table 2: Difference in C4 and FGF19 AUC over 24 hours at steady state between EYP001a and placebo treatments, in fed condition in healthy volunteers (* doses being assessed in phase 2a NASH trial)

Dosing regimen	C4 $\Delta AUC_{24,SS}$ $ng.h.ml^{-1}$	FGF19 $\Delta AUC_{24,SS}$ $pg.h.ml^{-1}$
100 mg QD morning	-105	504
200 mg QD* morning	-142	1508
100 mg BID*	-211	879
400 mg QD* morning	-177	3421
600 mg QD morning	-196	4760

Model Simulation

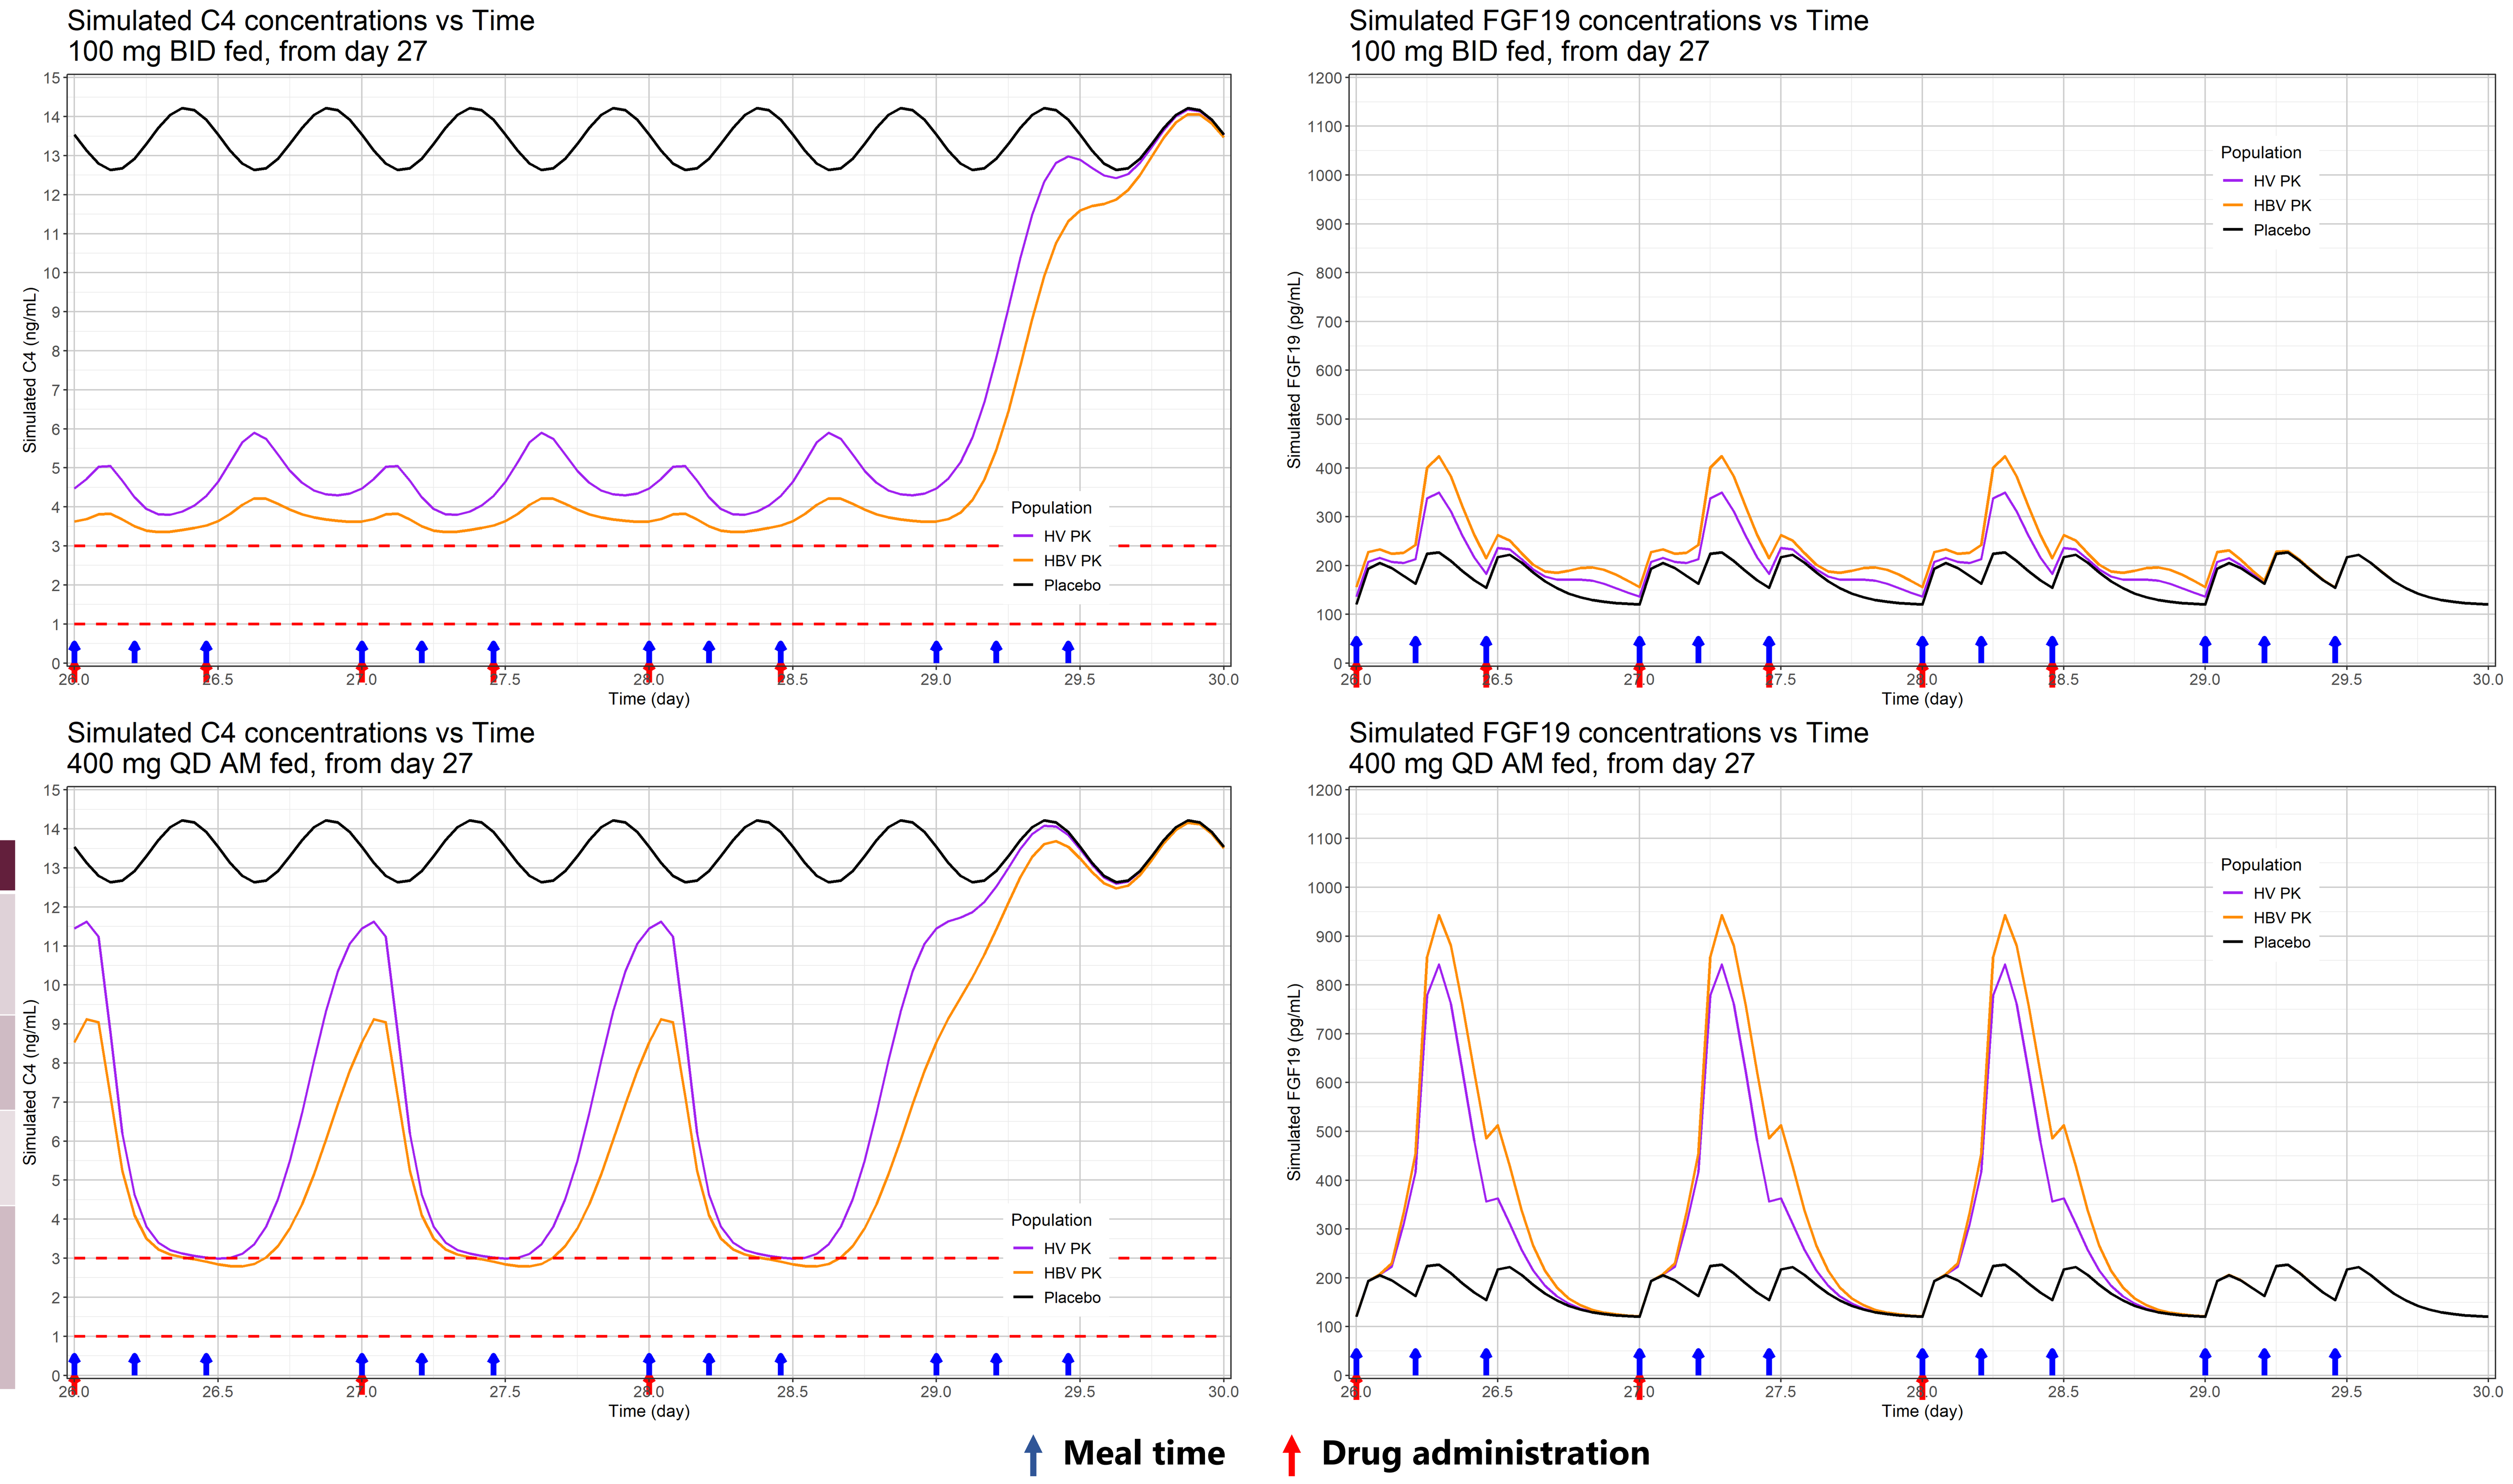
Simulated C4 concentrations vs Time
100 mg BID fed, from day 27
Simulated FGF19 concentrations vs Time
100 mg BID fed, from day 27
Simulated C4 concentrations vs Time
400 mg QD AM fed, from day 27
Simulated FGF19 concentrations vs Time
400 mg QD AM fed, from day 27
Simulated C4 (ng/mL)
Simulated FGF19 (pg/mL)
Time (day)
Population
HV PK
HBV PK
Placebo
Meal time
Drug administration

Figure 2: Simulations of C4 and FGF19 concentrations after repeated administration of EYP001a 100 mg BID (top) and 400 mg QD (bottom)

CONCLUSION

EYP001a, C4 and FGF19 concentrations were adequately described by the proposed approach and confirm the impact of EYP001a on FXR pathway.

Simulations showed that the maximal reduction of C4 was reached with 100 mg twice daily (BID) dosing and was not further decreased with higher doses. However, increasing the dose, increased the duration of the effect on C4. Also increasing EYP001a dose increased FGF19 concentrations, without a substantial additional effect on C4 concentrations.

This modelling was used to support the design of an ongoing phase 2a NASH trial. The model will be expanded to safety aspects to assess the benefice-risk balance and better identify the recommended dosing regimen.

REFERENCES

[1] Radreau, P et al, FASEB J. 30, 2016 Sep;30(9):3146-54.
[2] Sheiner, L.B et al, CPT, 1979 25(3): p. 358-71.
[3] Jacqmin, P et al, JPKPD. 2007; 34(1):57-85
[4] Lundasen, T et al. J Intern Med 2006, 260(6): p. 530-6.

DISCLOSURES

Pierrillas P. and Laveille C. are respectively employee and director of Calvagone
Scalfaro P. and André P. are consultants of ENYO Pharma SA
Darteil R., Barzic N., and Couchoux H. are employees of ENYO Pharma SA
Vonderscher J. is director and shareholder of ENYO Pharma, stock shareholder at Obseva SA and Hoffman La Roche AG, and board member at Obseva SA, Inatherys SAS, Step Pharma SAS and Inotrem SA.

CONTACT INFORMATION

Pierrillas Philippe: philippe.pierrillas@calvagone.com

AASLD AMERICAN ASSOCIATION FOR THE STUDY OF LIVER DISEASES

CALVAGONE

ENYO PHARMA