A novel small molecule modulating the mitochondrial NEET (CISD) proteins activity improves inflammation and fibrosis in a diet-induced model of non-alcoholic steatohepatitis

Stéphanie Le Corre[1], Laetitia Lines[1], Diane Sampson[1], Ola Karmi[2], Henri-Baptiste Marjault[2], Rodrigue Rossignol[3], Nivea Dias Amoedo[3], Rachel Nechushtai[2], Raphaël Darteil[1], Jacky Vonderscher[1,] Benoît de Chassey[1,] and Eric Meldrum[1]
[1]ENYO Pharma, Lyon, France
[2]The Alexander Silberman Life Science Institute, The Hebrew University of Jerusalem, Jerusalem, Israel
[3]Cellomet, Bordeaux, France

INTRODUCTION

Non-alcoholic steatohepatitis (NASH) is a disease characterized by excessive fat accumulation, inflammation, liver cell ballooning and fibrosis in the liver with periportal inflammation having been identified as a marker of disease severity[1]. Here, we present a novel chemistry that regulates the function of 3 mitochondrial proteins called the NEET proteins to target the periportal region of the liver. These proteins play an important role in mitochondrial Fe/ROS homeostasis and the regulation of mitochondrial metabolism.

AIM

Previous literature reports have highlighted the therapeutic importance of NEET proteins in metabolic disease[2,3], so we assessed the possible effects of ENYO's lead NEET protein modulator on the pathology associated with a diet-induced obesity model of NASH in mice and compared those effects with OCA treatment.

METHOD

In vitro experiments

• UV-vis spectroscopy was used to observe the 2Fe-2S cluster stability

• Effects on mitochondrial activity were assessed by evaluating (1) mitochondrial respiration by high-resolution respirometry and (2) the activity of electron transport chain complexes by standardized enzymology assays

• The anti-inflammatory effect has been assessed in a NFKB reporter cell line and in Kupffer cells (IL6 ELISA)

In vivo experiments

• Mice were fed with an AMLN diet (high in fat, fructose and cholesterol) for 30 weeks and then treated by oral administration, twice daily with compounds for 8 weeks. Effects were determined by comparing liver biopsy pathology at baseline and upon study completion. For comparison, a mouse group was also administered OCA daily.

RESULTS

In vitro

❶ Cluster assay stabilization by ENYO's molecule

Molecule	Time to achieve 50% loss of cluster	Residual cluster bound
dEF	55'	5%
DMSO	100'	0.70%

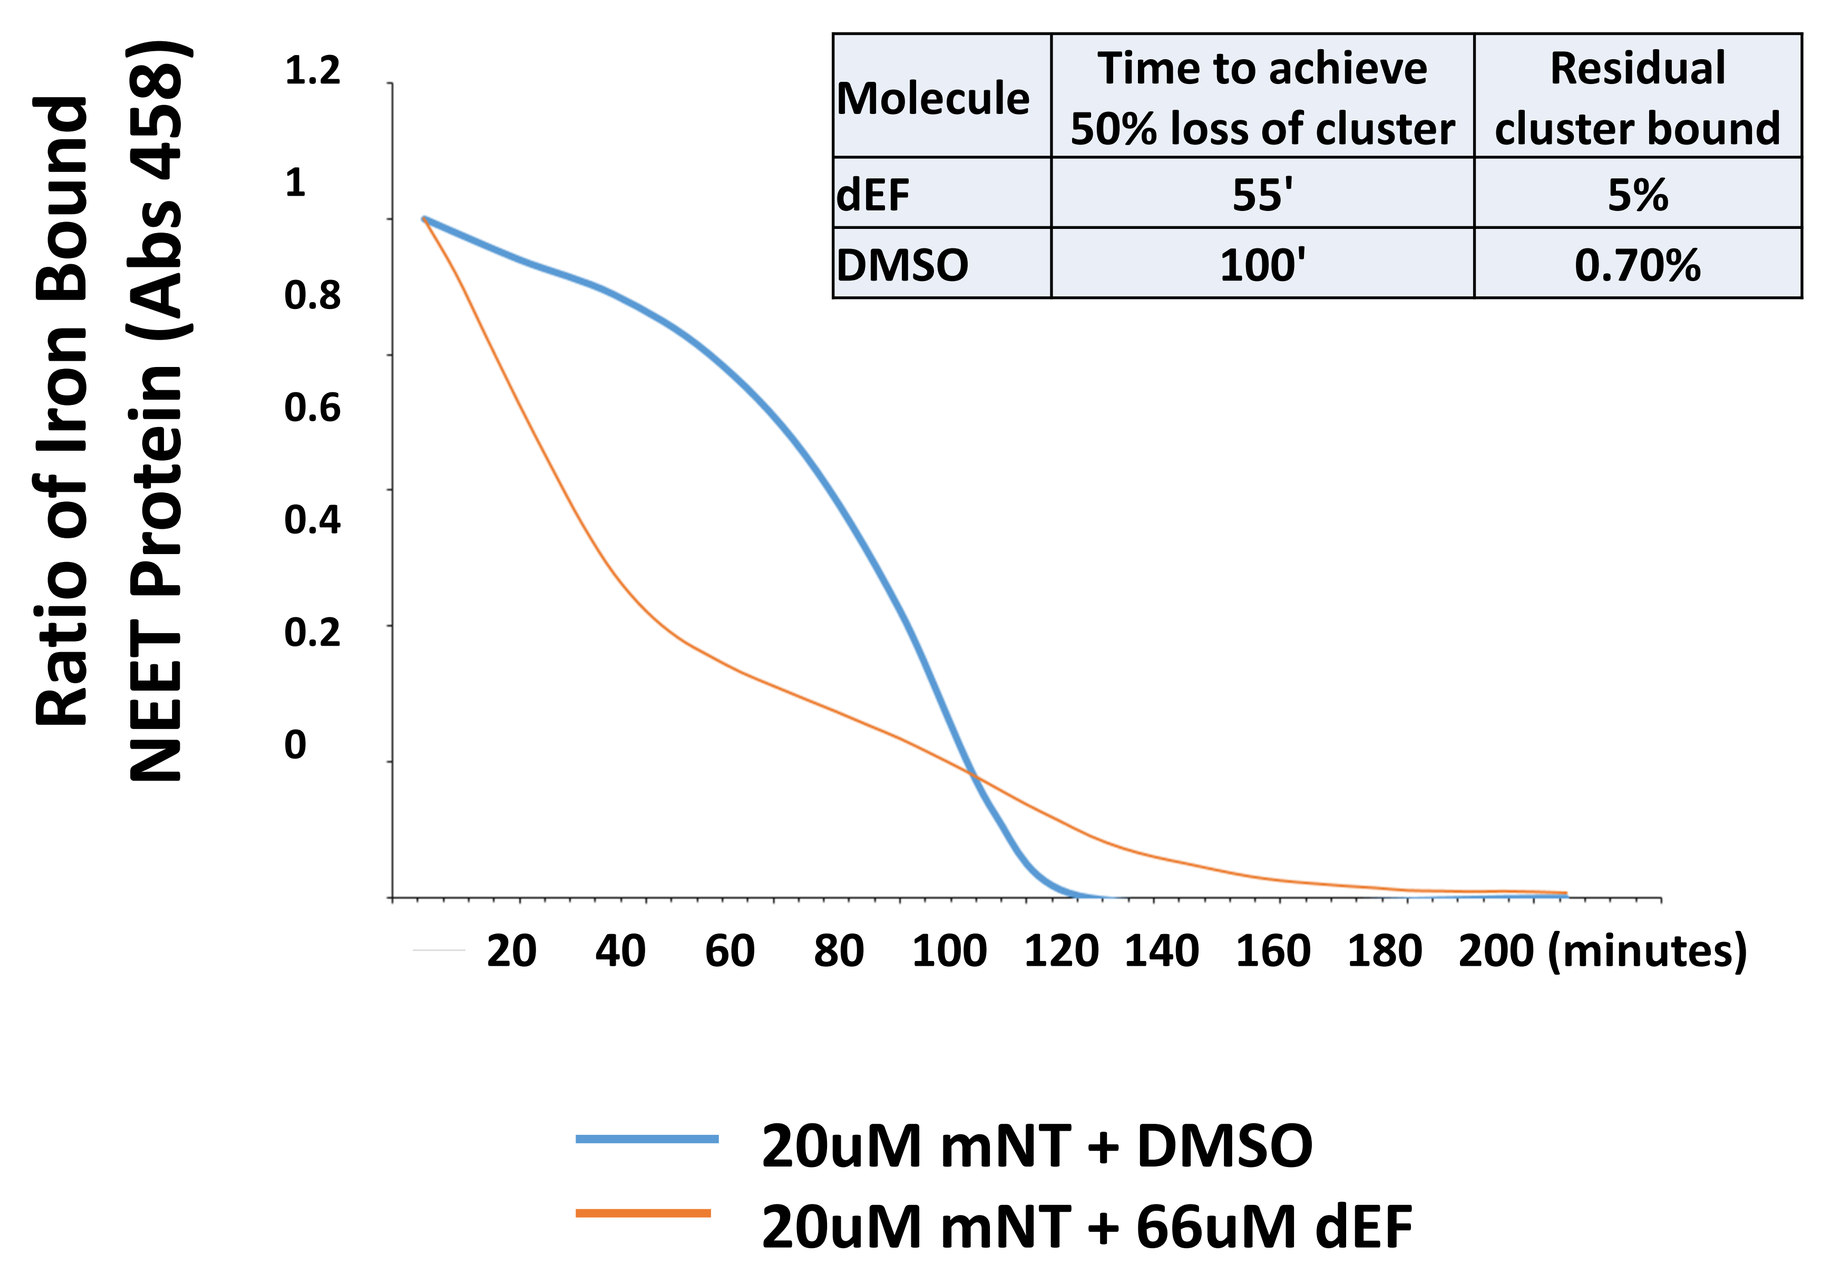
Ratio of Iron Bound NEET Protein (Abs 458)
1.2
1
0.8
0.6
0.4
0.2
0
20 40 60 80 100 120 140 160 180 200 (minutes)
20uM mNT + DMSO
20uM mNT + 66uM dEF

• UV-vis spectroscopy was used to assess the 2Fe-2S cluster stability of NAF-1 or mNT or MiNT in the absence and presence of ENYO's molecule (dEF).

• Our molecule destabilizes the 2Fe-2S cluster of the 3 NEET proteins. As an example we show here the result on mNT.

❷ Modulation of Mitochondrial Respiration

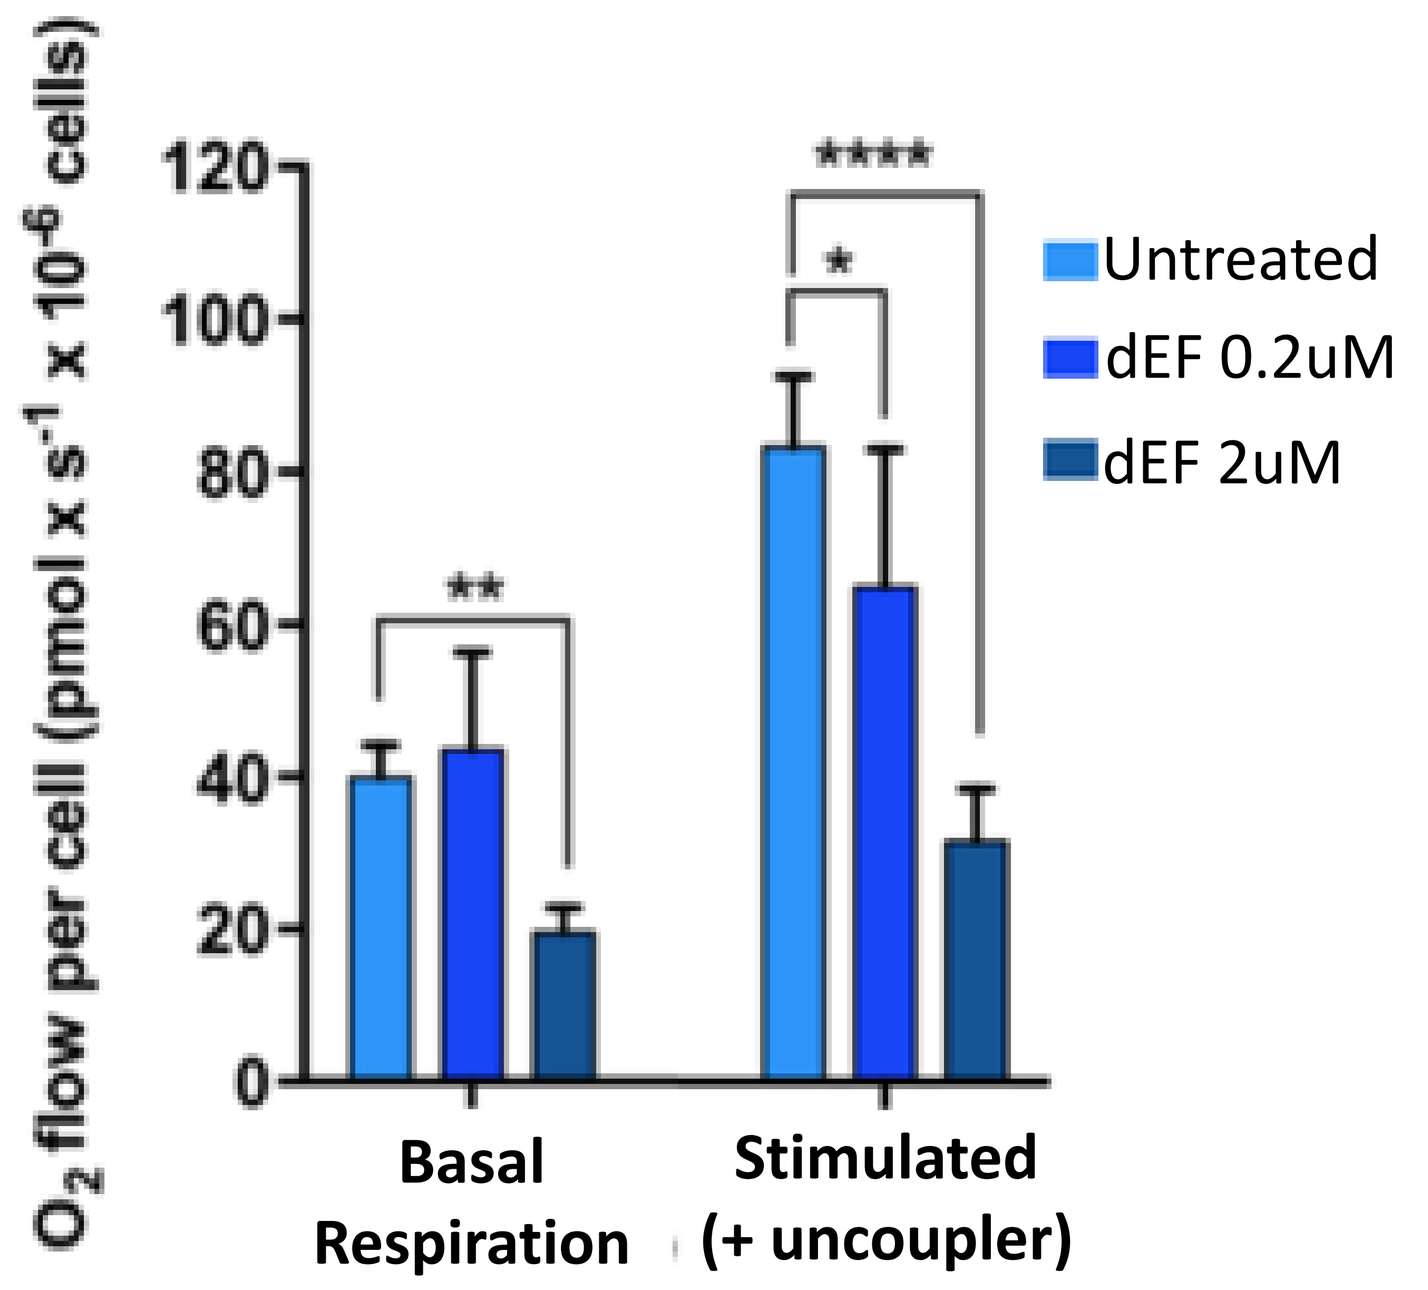
O2 flow per cell (pmol x s-1 x 10-6 cells)
120
100
80
60
40
20
0
**
*

Untreated
dEF 0.2uM
dEF 2uM
Basal Respiration
Stimulated (+ uncoupler)

• High-resolution respirometry studies has been performed on A549 cells (cellular oxygen consumption in a closed-chamber system).

• An inhibition of routine respiration and uncoupled maximal respiration (+CCCP) is observed.

• ENYO's molecule inhibits the ability of a cell to enhance oxidative phosphorylation upon stimulation (as induced by metabolic, oxidative or other cellular stress).

❸ Inhibition of Electron Transport Chain Complex 1

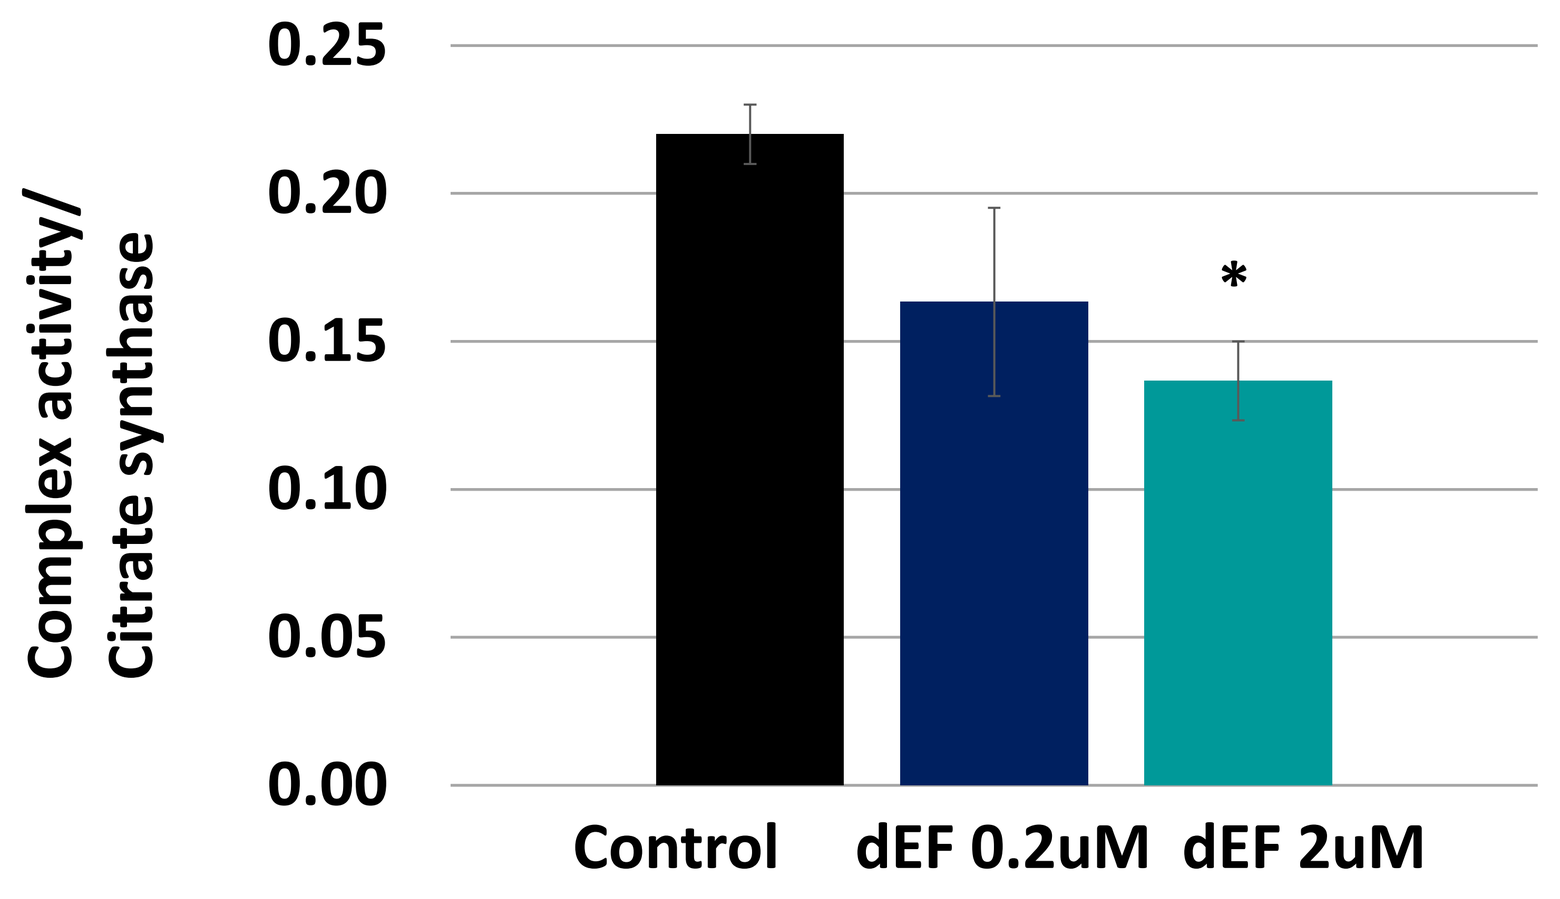
Complex activity/ Citrate synthase
0.25
0.20
0.15
0.10
0.05
0.00
*
Control
dEF 0.2uM
dEF 2uM

• Rotenone-sensitive NADH-ubiquinone oxido-reductase activity rate is measured spectrophotometrically on cell homogenate and complex I specific function is verified using rotenone, a specific Complex 1 blocker. A significant inhibition of respiratory chain complex 1 activity was measured.

❹ ENYO's molecule reduces inflammation *in vitro*

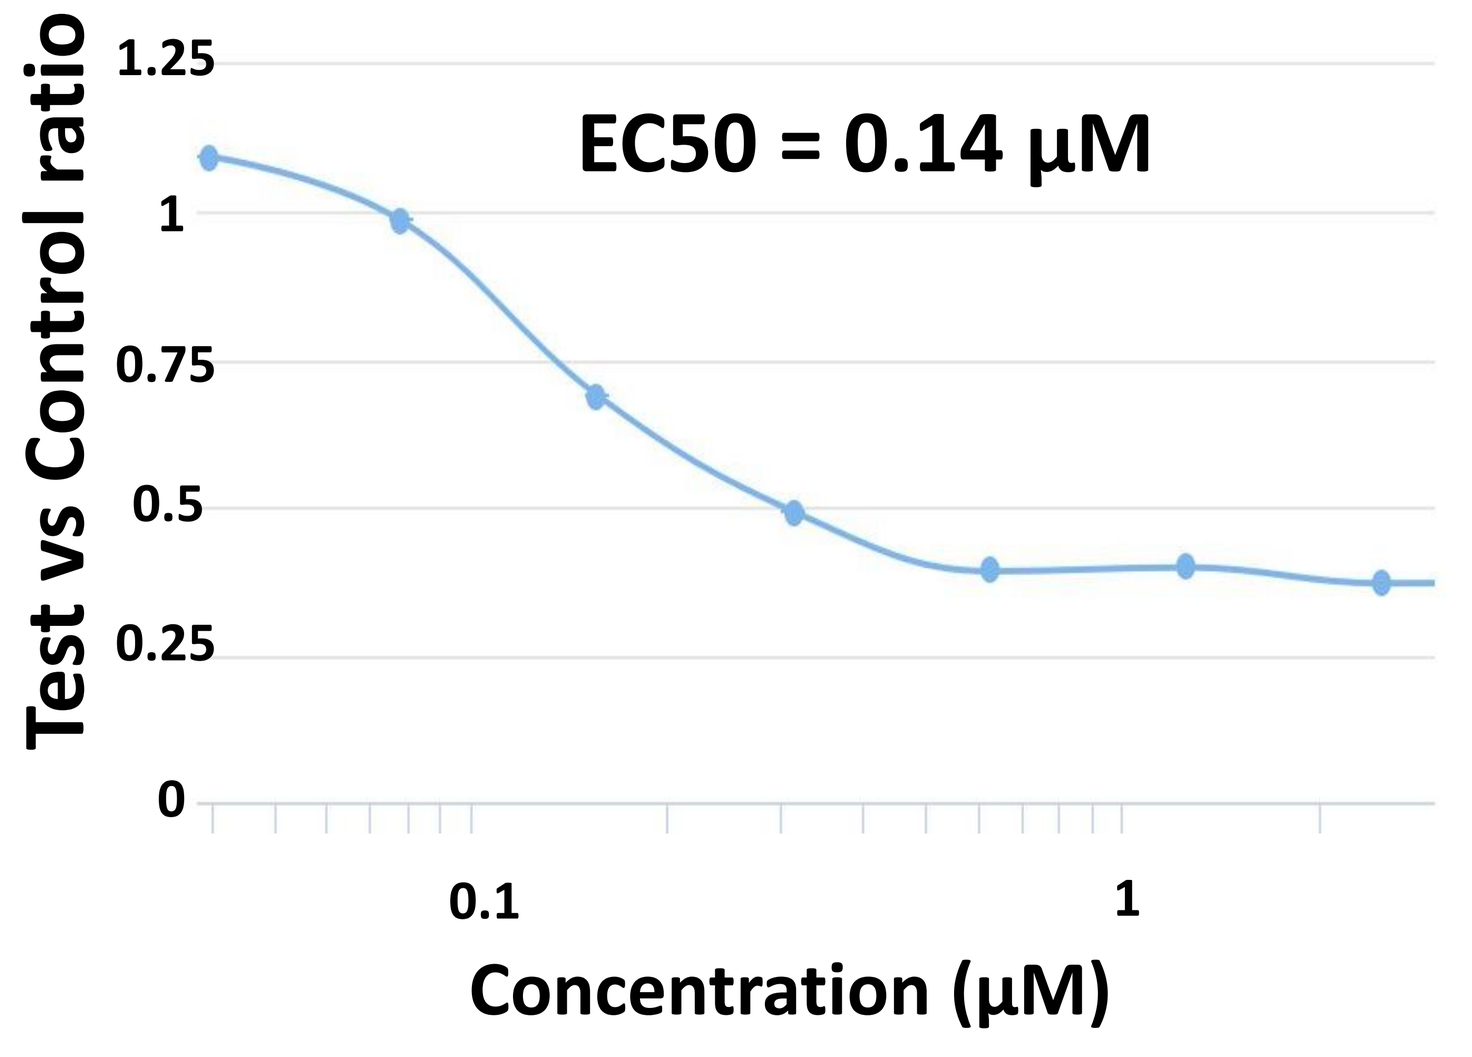
EC50 = 0.14 µM
Test vs Control ratio
1.25
1
0.75
0.5
0.25
0
0.1
1
Concentration (µM)

•The NFKB reporter cell line has been induced by TNFα for 6 h

• The signal is detected using a NanoGlo Luciferase Assay System

• ENYO's molecule inhibits NFKB activation by TNFα

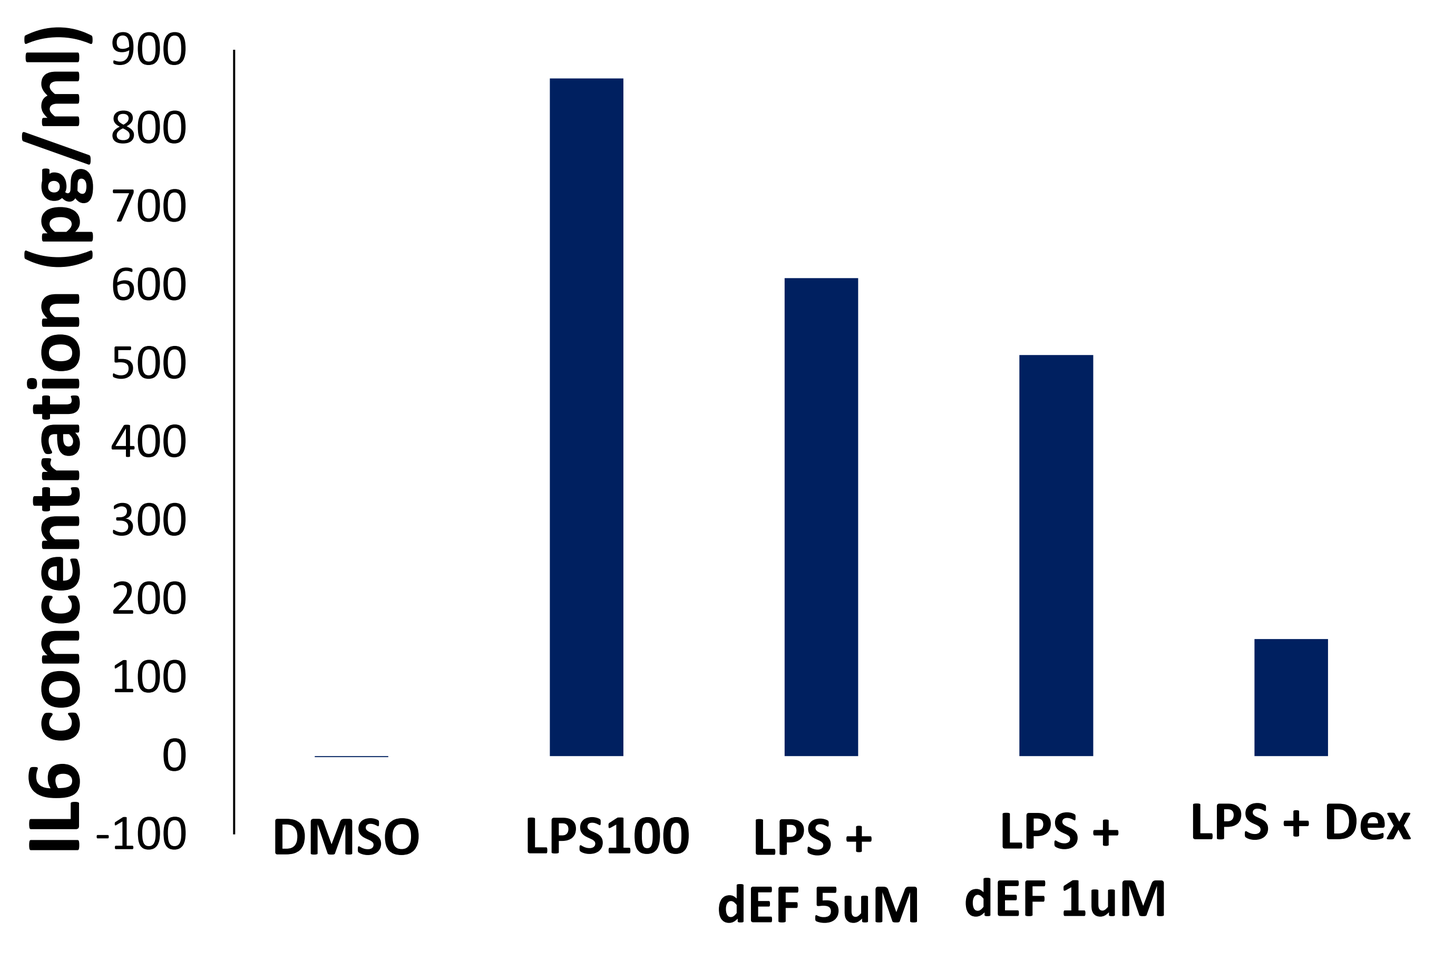
IL6 concentration (pg/ml)
900
800
700
600
500
400
300
200
100
0
-100
DMSO
LPS100
LPS + dEF 5uM
LPS + dEF 1uM
LPS + Dex

• Kupffer cells are induced by LPS for 24h. IL6 production is detected in culture supernatant with an ELISA assay

• Dexamethasone has been used as a positive control

• ENYO's molecule decreases IL6 production in induced Kupffer cells. Of note, the maximum effect is reached at the lowest concentration tested, 1uM.

In vivo

❺ Reduction of liver Inflammation and Fibrosis *in vivo*

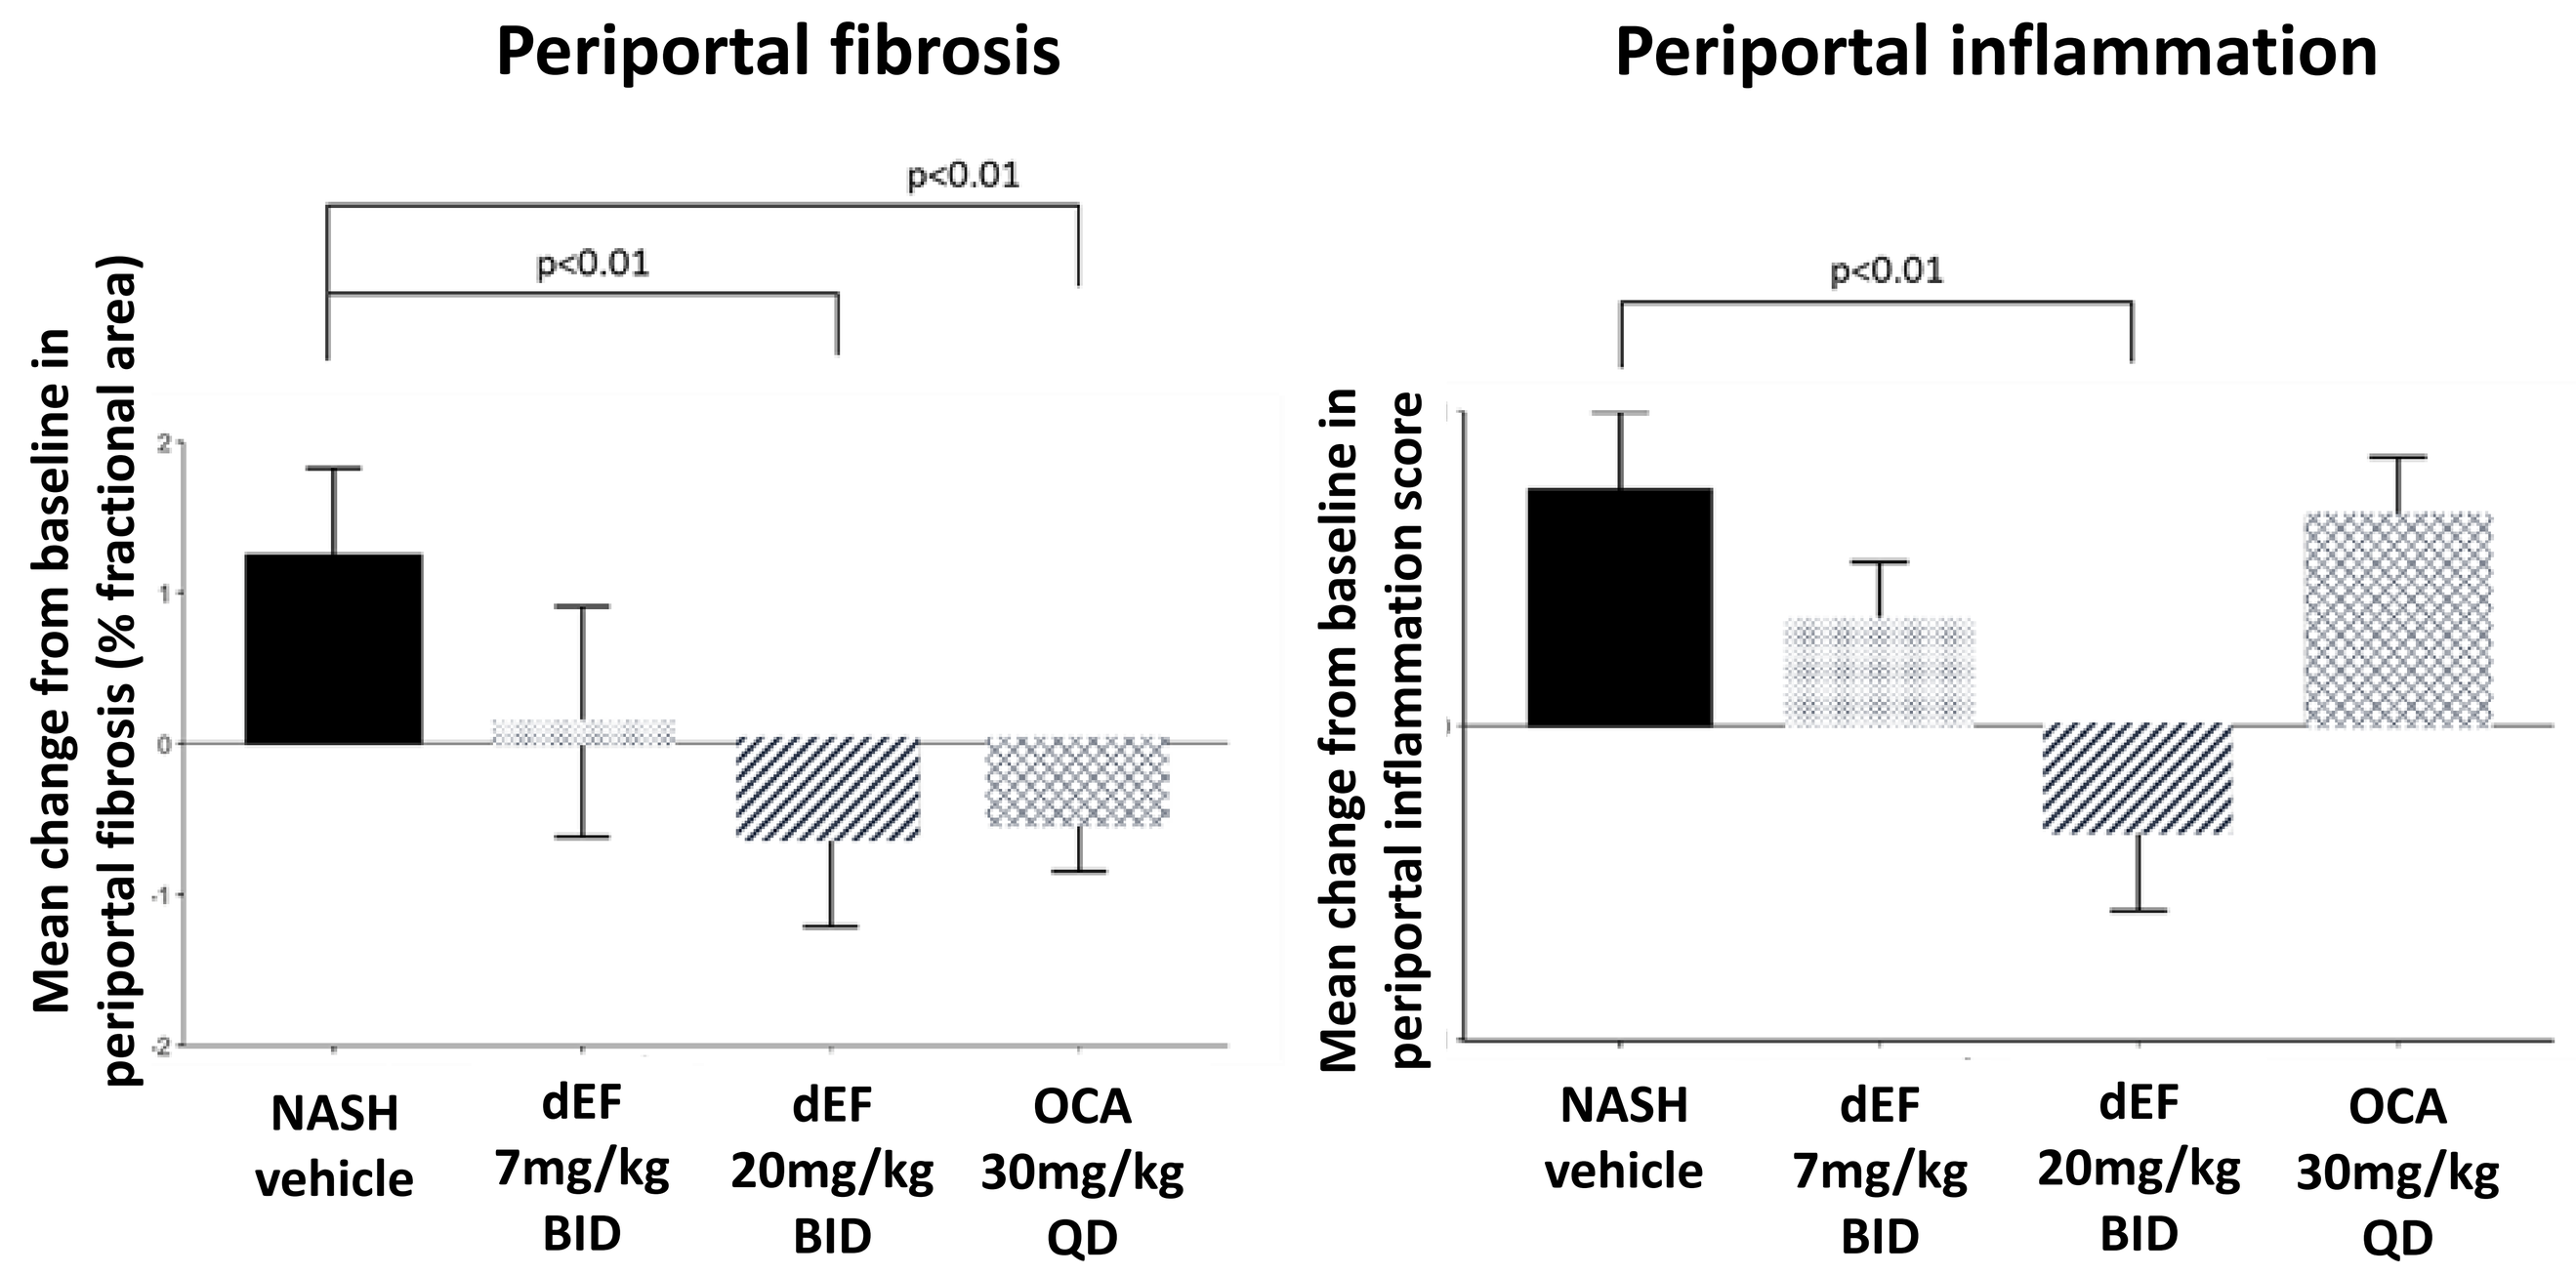
Periportal fibrosis
p<0.01
p<0.01
Mean change from baseline in periportal fibrosis (% fractional area)
NASH vehicle
dEF 7mg/kg BID
dEF 20mg/kg BID
OCA 30mg/kg QD
Periportal inflammation
p<0.01
Mean change from baseline in periportal inflammation score
NASH vehicle
dEF 7mg/kg BID
dEF 20mg/kg BID
OCA 30mg/kg QD

• Inflammation is graded by number of H&E positive inflammatory foci per field.

• Fibrosis is graded by image analysis and quantification of PSR positive staining in the region 100 µm from the portal tract.

• Therapeutic use of ENYO's molecule resolves periportal inflammation and periportal fibrosis following 8 weeks of treatment in a NASH mouse model.

CONCLUSIONS

ENYO's lead molecule is well tolerated *in vivo* and resolves periportal inflammation and periportal fibrosis in a NASH mouse model.

In the liver, improved pathology is biased to the hepatic periportal zone which is perfused with blood with highest oxygen content[4]. We propose hepatocytes, pro-inflammatory & pro-fibrotic cells in the well-perfused periportal zone are most sensitive to ENYO's molecule by virtue of their bioenergetic predominant oxydative (OxPhos$^+$) status and metabolic inflexibility.

ENYO's lead series offers a novel approach to regulate the activity of a family of proteins of critical importance in mitochondrial biology. It may offer new perspectives in the treatment of NASH and other diseases with an important inflammatory component.

REFERENCES

1- Liver Fibrosis, but no Other Histologic Features, Associates with Long-term Outcomes of Patients With Nonalcoholic Fatty Liver Disease *(Angulo et al 2015, Gastroenterology, 149(2) 389-397)*

2- NAF-1 inhibitor improves diabetic endpoints in db/db mice *(Takahashi et al 2015, J.Pharmacol. Exp. Ther.* ***352*** *p338)*

3- NAF-1 haploinsufficiency results in NASH-like pathology *(Shen et al 2017 Cell Reports,* ***21*** *p2198*

4- Spatial heterogeneity in the mammalian liver *(Shani Ben-Moshe and Shalev Itzkovitz 2019, Nature reviews)*

CONTACTS

Stéphanie Le Corre: slc@enyopharma.com

Raphael Darteil: rda@enyopharma.com

Eric Meldrum: ecm@enyopharma.com